317

Impact of Prior Chronic Inflammatory Demyelinating Polyradiculoneuropathy Treatments in the ADHERE Trial: ***Post Hoc*** **Analyses**

Jeffrey A. Allen,[1] Jie Lin,[2] Mark Stettner,[3] Jeffrey T. Guptill,[4,5] Geoffrey Istas,[5] Arne De Roeck,[5] Satoshi Kuwabara,[6] Giuseppe Lauria,[7,8] Luis Querol,[9,10] Silvia Rossi,[11] Niraja Suresh,[12] Chafic Karam,[13] Thomas Skripuletz,[14] Simon Rinaldi,[15] Andoni Echaniz-Laguna,[16] Channa Hewamadduma,[17,18] Benjamin Van Hoorick,[5] Ryo Yamasaki,[19] Pieter A. van Doorn,[20] Richard A. Lewis[21]

[1]University of Minnesota, Minneapolis, MN, USA; [2]Huashan Hospital, Fudan University, Shanghai, China; [3]Center for Translational Neuro- and Behavioral Sciences (C-TNBS), University Medicine Essen, Essen, Germany; [4]School of Medicine, Duke University, Durham, NC, USA; [5]argenx, Ghent, Belgium; [6]Graduate School of Medicine, Chiba University, Chiba, Japan; [7]IRCCS Foundation "Carlo Besta" Neurological Institute, Milan, Italy; [8]University of Milan, Milan, Italy; [9]Department of Neurology, Neuromuscular Diseases Unit, Hospital de La Santa Creu I Sant Pau, Universitat Autònoma de Barcelona, Barcelona, Spain; [10]Centro De Investigación Biomédica en Red en Enfermedades Raras (CIBERER), Madrid, Spain; [11]argenx, Milan, Italy; [12]Lakeland Regional Health, Lakeland, FL, USA; [13]University of Pennsylvania, Philadelphia, PA, USA; [14]Hannover Medical School, Hanover, Germany; [15]University of Oxford, Oxford, UK; [16]French National Reference Center for Rare Neuropathies (CERAMIC), Bicêtre University Hospital, Le Kremlin-Bicêtre, France; [17]Academic Neuromuscular Unit, Sheffield Teaching Hospitals Foundation NHS Trust, Sheffield, UK; [18]Sheffield Institute for Translational Neuroscience (SITRAN), University of Sheffield, Sheffield, UK; [19]Kyushu University Hospital, Fukuoka, Japan; [20]Erasmus University Medical Center, Rotterdam, the Netherlands; [21]Cedars-Sinai Medical Center, Los Angeles, CA, USA

BACKGROUND | METHODS

- CIDP is a rare, severe, progressive immune -mediated disease leading to disability due to proximal and/or distal weakness and sensory disturbance[1,2]
- Efgartigimod is a human IgG antibody Fc fragment that blocks the neonatal Fc receptor[3]
- Efgartigimod outcompetes endogenous IgG, decreases IgG recycling, promotes lysosomal degradation of IgG, and reduces IgG levels without impacting IgG production[3–8]
- In the ADHERE trial (NCT04281472), efgartigimod PH20 SC (co-formulated with recombinant human hyaluronidase PH20)[9] reduced relapse risk, including in those who received prior treatment, and was well tolerated in participants with CIDP[6]

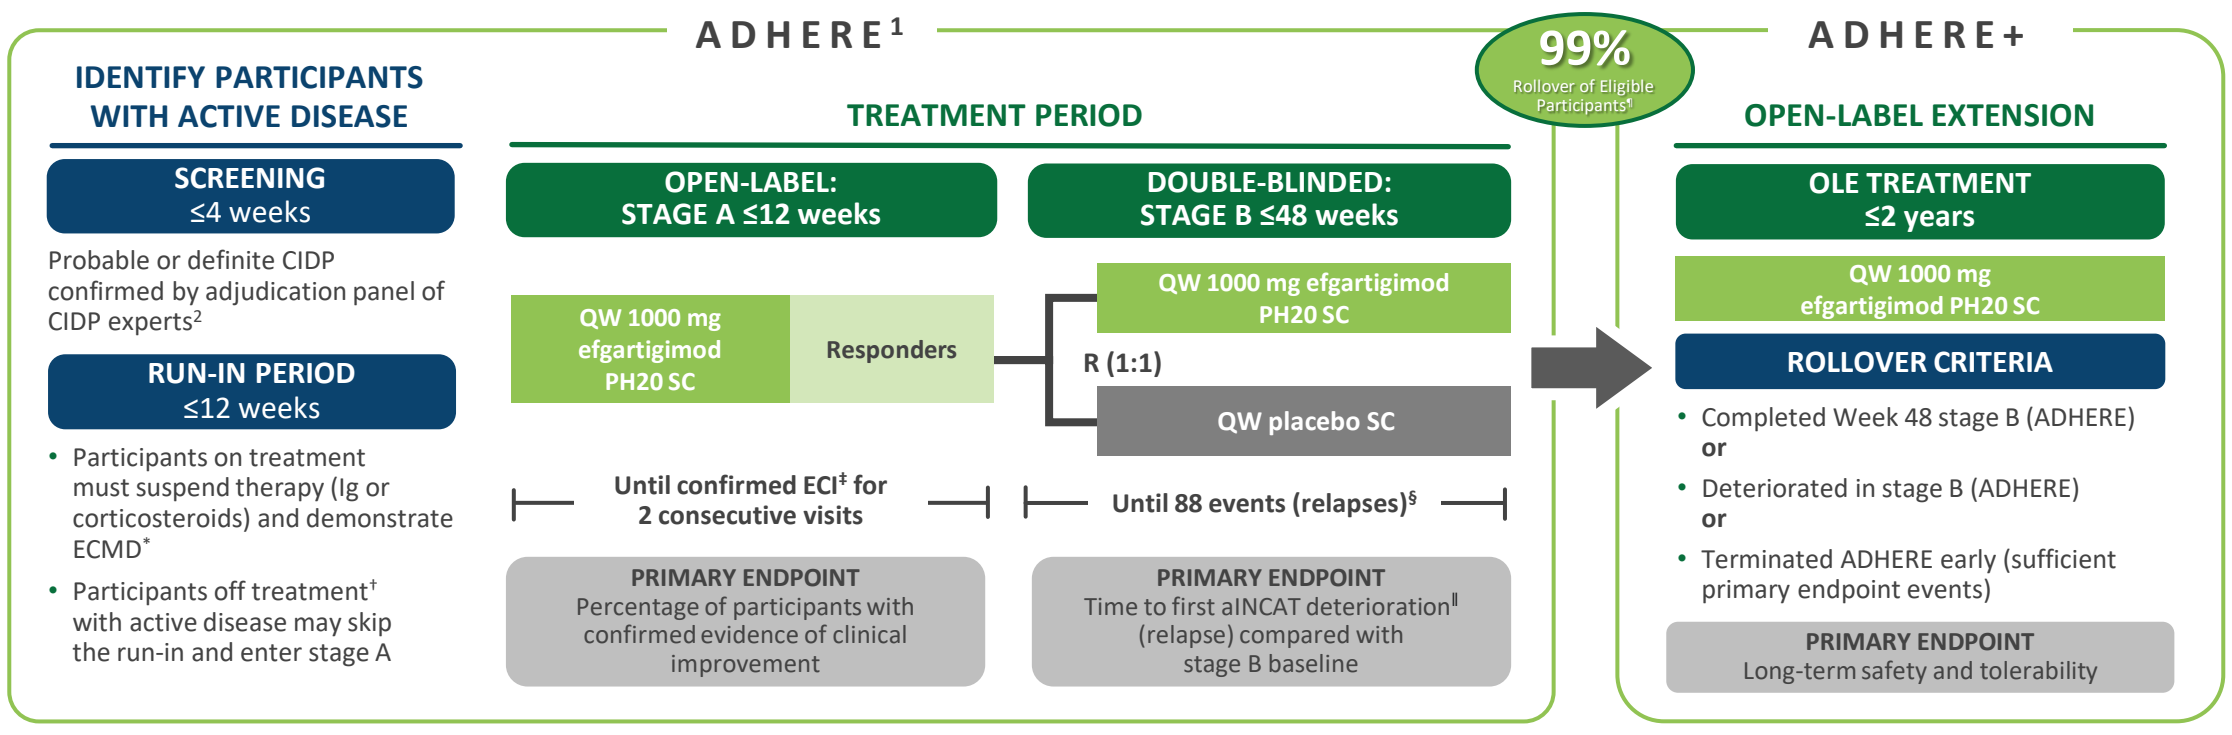

ADHERE[1]
IDENTIFY PARTICIPANTS WITH ACTIVE DISEASE
SCREENING ≤4 weeks
Probable or definite CIDP confirmed by adjudication panel of CIDP experts[2]
RUN-IN PERIOD ≤12 weeks
Participants on treatment must suspend therapy (Ig or corticosteroids) and demonstrate ECMD*
Participants off treatment† with active disease may skip the run-in and enter stage A
TREATMENT PERIOD
OPEN-LABEL: STAGE A ≤12 weeks
QW 1000 mg efgartigimod PH20 SC
Responders
DOUBLE-BLINDED: STAGE B ≤48 weeks
QW 1000 mg efgartigimod PH20 SC
R (1:1)
QW placebo SC
Until confirmed ECI‡ for 2 consecutive visits
Until 88 events (relapses)§
PRIMARY ENDPOINT
Percentage of participants with confirmed evidence of clinical improvement
PRIMARY ENDPOINT
Time to first aINCAT deterioration‖ (relapse) compared with stage B baseline
99%
Rollover of Eligible Participants¶
ADHERE+
OPEN-LABEL EXTENSION
OLE TREATMENT ≤2 years
QW 1000 mg efgartigimod PH20 SC
ROLLOVER CRITERIA
Completed Week 48 stage B (ADHERE) or
Deteriorated in stage B (ADHERE) or
Terminated ADHERE early (sufficient primary endpoint events)
PRIMARY ENDPOINT
Long-term safety and tolerability

*ECMD was defined as an aINCAT increase of ≥1 points, an I-RODS decrease of ≥4 points (centile metric), or a grip strength decrease of ≥8 kPa. †Off treatment was defined as participants who had never received CIDP treatment (treatment naïve) or who had not received CIDP treatment (corticosteroids, IVIg, or SCIg) within 6 months of trial entry. ‡ECI was defined as a clinical improvement on the parameters that the participant worsened in during run-in (≥4-point increase in I-RODS and/or ≥8-kPa increase in mean grip strength) or clinical improvement (≥1-point decrease) in aINCAT. ECI was confirmed after these criteria were met after 4 injections and 2 consecutive visits. §The primary endpoint was assessed once 88 total relapses or events were achieved in stage B and was based on the HR for the time to first aINCAT deterioration (ie, relapse). ‖aINCAT deterioration was defined as a ≥1-point increase in aINCAT score compared with stage B baseline, which was confirmed at a consecutive visit after the first 1-point increase in aINCAT or not confirmed for participants with ≥2-point increase in aINCAT compared with stage B baseline. ¶n=228/229. 229 participants enrolled in ADHERE+, including 3 participants who inadvertently rolled over without meeting per-protocol inclusion criteria. The safety population for ADHERE+ included 228 participants who received ≥1 dose of efgartigimod PH20 SC in the OLE, as 1 participant discontinued before receiving the first dose of efgartigimod PH20 SC.

OBJECTIVE

In this *post hoc* analysis, we report outcomes with efgartigimod PH20 SC by prior treatment received for CIDP

RESULTS

Baseline Characteristics

- Baseline characteristics have previously been described (Allen, 2024)[6]
- 322 participants entered stage A:
 - 63 (19.6%) had received prior corticosteroids
 - 165 (51.2%) had received prior IVIg/SCIg
 - 94 (29.2%) were off treatment before entry into ADHERE
- Mean time since diagnosis was similar in the corticosteroids and off treatment subgroups (3.8 and 3.9 years), and longer in the IVIg/SCIg subgroup (5.8 years)
- Participants in the efgartigimod PH20 SC and placebo arms were stratified to enter stage B by most recent prior treatment and aINCAT score change during stage A (**Table 1**)

TABLE 1 Prior CIDP Treatment in Stage B

	Efgartigimod PH20 SC (N=111)	Placebo SC (N=110)	Total (N=221)
Prior CIDP therapy, n (%)			
Corticosteroids	26 (23.4)	24 (21.8)	50 (22.6)
IVIg/SCIg	49 (44.1)	47 (42.7)	96 (43.4)
Off treatment	36 (32.4)	39 (35.5)	75 (33.9)

- 19 participants in the IVIg/SCIg subgroup experienced the adverse event of CIDP worsening (n=17), muscle weakness (n=1), or quadriparesis (n=1); of these, 17 recovered, 1 partially recovered and 1 died during follow-up (deemed unlikely related to treatment)
- Study discontinuation rates were higher in the prior IVIg/SCIg subgroup than in the prior corticosteroid or off treatment subgroups
 - A quarter of these IVIg/SCIg participants received <4 efgartigimod PH20 SC injections (minimum treatment required to achieve maximal IgG reduction) (**Table 2**)
 - Not all participants who withdrew from stage A experienced deterioration on aINCAT (**Table 2, Figure 1**)

TABLE 2 Reasons for Study/Treatment Discontinuation by Prior CIDP Medication

	Corticosteroid (N=63)	IVIg/SCIg (N=165)	Off Treatment (N=94)
Participants who discontinued stage A treatment	8 (12.7)	55 (33.3)	8 (8.5)
Adverse event	2 (3.2)	18 (10.9)	0
Death	0	1 (0.6)	0
Lack of efficacy	3 (4.8)	21 (12.7)	4 (4.3)
Lost to follow-up	0	1 (0.6)	1 (1.1)
Noncompliance with study drug	1 (1.6)	0	0
Physician decision	0	1 (0.6)	0
Prohibited medications	0	2 (1.2)	0
Withdrawal by subject	1 (1.6)	8 (4.8)	1 (1.1)
Other	1 (1.6)	3 (1.8)	2 (2.1)
Participants with all events required for primary analysis achieved	4 (6.3)	11 (6.7)	7 (7.4)
Participants who completed stage A treatment	51 (81.0)	99 (60.0)	79 (84.0)
Participants who received <4 injections and discontinued from the trial	2 (3.2)	42 (25.5)	3 (3.2)
All events required for primary analysis achieved	0	4 (2.4)	3 (3.2)
Any other reason	2 (3.2)	38 (23.0)	0
Adverse event	1 (1.6)	15 (9.1)	0
Death	0	1 (0.6)	0
Lack of efficacy before end of stage A	1 (1.6)	12 (7.3)	0
Lost to follow-up	0	1 (0.6)	0
Noncompliance with study drug	0	0	0
Physician decision	0	1 (0.6)	0
Prohibited medications	0	1 (0.6)	0
Withdrawal by subject	0	5 (3.0)	0
Other	0	2 (1.2)	0

TABLE 3 Change in aINCAT From Run-In Baseline – Pretreated Population*

	Prior Corticosteroid (N=50)	Prior IVIg/SCIg (N=96)	Pretreated Participants* (N=146)
Run-in baseline			
Mean (95% CI)	4.14 (3.68, 4.60)	3.86 (3.62, 4.11)	3.96 (3.73, 4.18)
Change/baseline (95% CI)	—	—	—
Stage A baseline			
Mean (95% CI)	4.68 (4.16, 5.20)	4.43 (4.10, 4.75)	4.51 (4.24, 4.79)
Change/baseline (95% CI)	0.54 (0.32, 0.76)	0.56 (0.36, 0.76)	0.55 (0.40, 0.70)
Stage B baseline			
Mean (95% CI)	3.55 (3.25, 4.07)	2.74 (2.44, 3.04)	3.05 (2.80, 3.31)
Change/baseline (95% CI)	−0.48 (−0.79, −0.17)	−1.12 (−1.44, −0.81)	−0.9 (−1.14, −0.67)
Stage B last assessment			
Mean (95% CI)	4.04 (3.51, 4.57)	3.54 (3.12, 3.96)	3.71 (3.38, 4.04)
Change/baseline (95% CI)	−0.1 (−0.55, 0.35)	−0.32 (−0.76, 0.11)	−0.25 (−1.57, −0.07)
Week 12 ADHERE+			
Mean (95% CI)	3.43 (2.95, 3.67)	2.35 (2.01, 2.69)	2.71 (2.42, 3.00)
Change/baseline (95% CI)	−0.71 (−1.18, −0.25)	−1.47 (−1.90, −1.04)	−1.22 (−1.54, −0.89)
Week 24 ADHERE+			
Mean (95% CI)	3.13 (2.59, 3.67)	2.11 (1.77, 2.46)	2.44 (2.14, 2.74)
Change/baseline (95% CI)	−1.05 (−1.63, −0.47)	−1.69 (−2.11, −1.27)	−1.49 (−1.73, −1.02)
Week 36 ADHERE+			
Mean (95% CI)	2.86 (2.23, 3.49)	2.40 (2.02, 2.77)	2.54 (2.22, 2.87)
Change/baseline (95% CI)	−1.39 (−2.01, −0.77)	−1.37 (−1.81, −0.93)	−1.38 (−1.73, −1.02)

*The pretreated population included participants that received either IVIg/SCIg or corticosteroids within 6 months prior to screening.

Efficacy

- All patients in ADHERE had to show deterioration (≥1 point on aINCAT, ≥4 points on centile metrics I-RODS, or ≥8 kPa on MGS) during a treatment-free run-in period and were already on a path of deterioration before starting efgartigimod in stage A. The percentage of patients with ≥1-point worsening on aINCAT from run-in baseline to stage A baseline (period capturing treatment withdrawal phase of the study) was 37.1% for prior corticosteroids, 41.7% for prior IVIg/SCIg, and 39.7% for the off-treatment group
- At stage A last assessment, the percentage of patients with a ≥1-point worsening on aINCAT compared with run-in baseline was lower than at stage A baseline in all prior treatment groups (**Figure 1**)
- Stage A ECI response rates were 77.8% for corticosteroids, 58.8% for IVIg/SCIg, and 72.3% for off treatment. At stage A last assessment, the majority of patients in all groups had equal or better aINCAT scores compared with run-in baseline (**Figure 1**)
- During stage B, further improvement was observed. In the prior IVIg/SCIg-treated participants randomized to efgartigimod, 34/49 (69.4%) improved ≥1 aINCAT point and 20/49 (40.8%) ≥2 aINCAT points at stage B best improvement compared with run-in baseline

FIGURE 1 Change in aINCAT Scores From Run-In Baseline* to Stage A Last Assessment by Prior CIDP Medication Subgroups

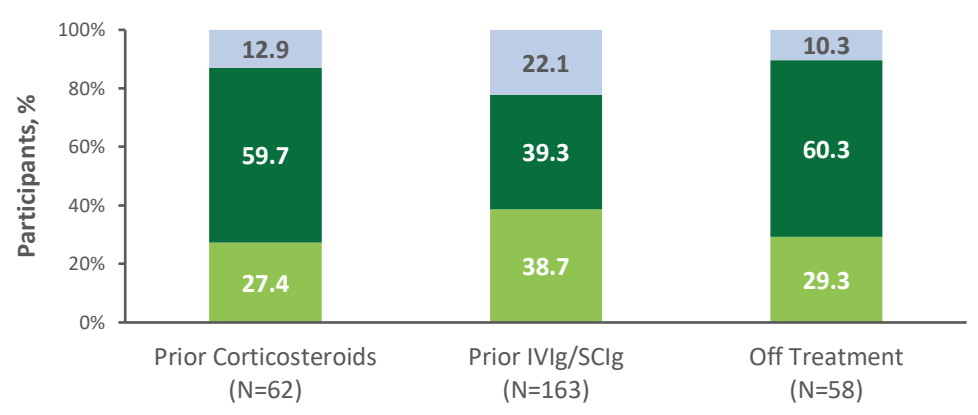

Improved (≥ −1 pt)
Remained stable (0 pt)
Worsened (≥1 pt)
100%
80%
60%
40%
20%
0%
Participants, %
12.9
59.7
27.4
22.1
39.3
38.7
10.3
60.3
29.3
Prior Corticosteroids (N=62)
Prior IVIg/SCIg (N=163)
Off Treatment (N=58)

*At stage A last assessment, 1.6% (corticosteroids), 16.0% (IVIg/SCIg), and 1.7% (off treatment) worsened ≥1 point on aINCAT compared with stage A baseline.

- Across all prior treatment subgroups, efgartigimod PH20 SC improved aINCAT scores through Week 36 of ADHERE+. These improvements corresponded with time on efgartigimod PH20 SC treatment (**Figure 2**)

FIGURE 2 Longitudinal Efficacy* by Prior CIDP Treatment Groups in Stage A Responders

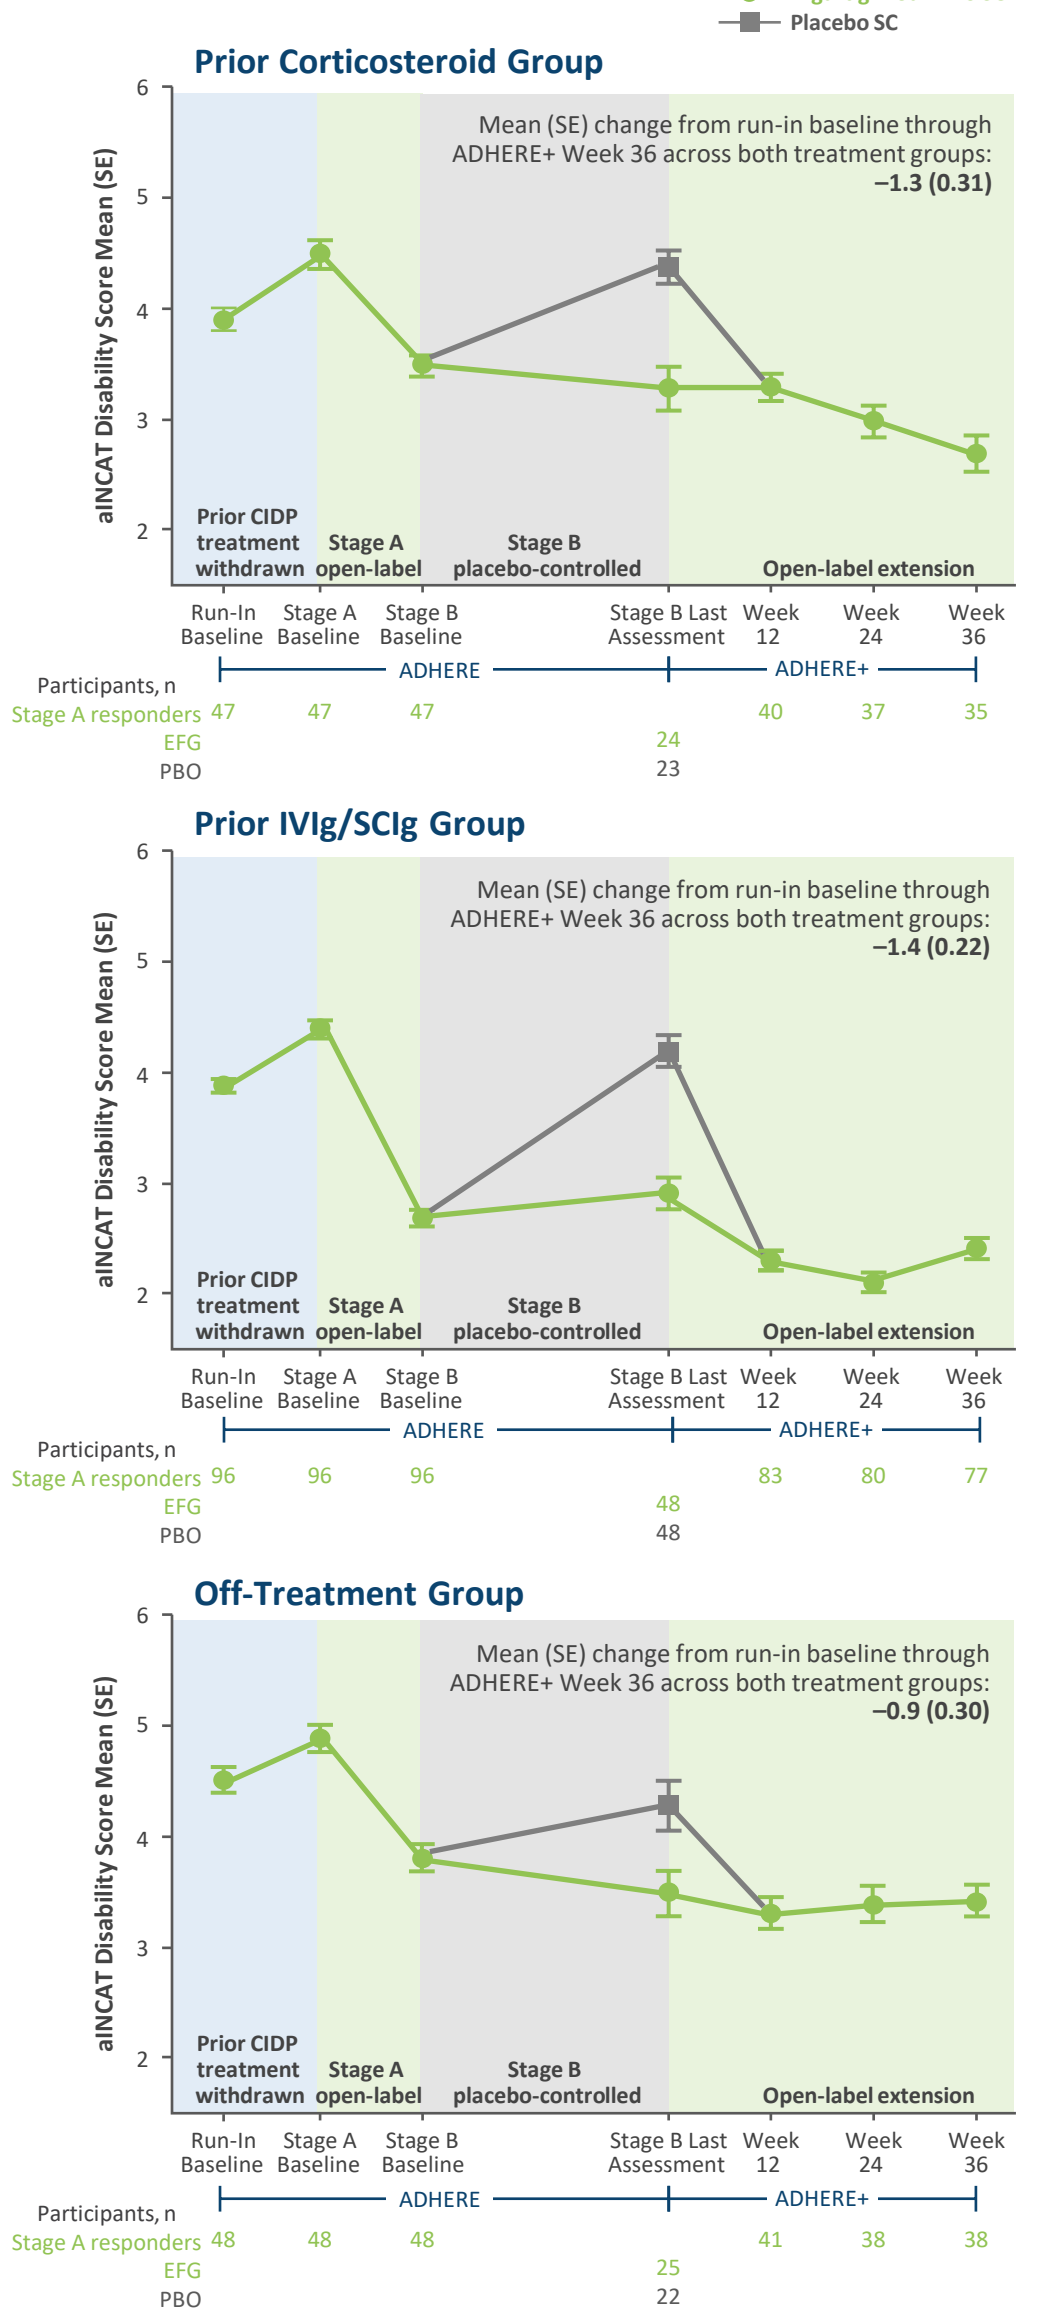

Efgartigimod PH20 SC
Placebo SC
Prior Corticosteroid Group
Mean (SE) change from run-in baseline through ADHERE+ Week 36 across both treatment groups: −1.3 (0.31)
aINCAT Disability Score Mean (SE)
Prior CIDP treatment withdrawn
Stage A open-label
Stage B placebo-controlled
Open-label extension
Run-In Baseline
Stage A Baseline
Stage B Baseline
Stage B Last Assessment
Week 12
Week 24
Week 36
ADHERE
ADHERE+
Participants, n
Stage A responders 47 47 47 40 37 35
EFG 24
PBO 23
Prior IVIg/SCIg Group
Mean (SE) change from run-in baseline through ADHERE+ Week 36 across both treatment groups: −1.4 (0.22)
Stage A responders 96 96 96 83 80 77
EFG 48
PBO 48
Off-Treatment Group
Mean (SE) change from run-in baseline through ADHERE+ Week 36 across both treatment groups: −0.9 (0.30)
Stage A responders 48 48 48 41 38 38
EFG 25
PBO 22

*Based on aINCAT Scores in the mITT population. 1-point difference in aINCAT is the minimal clinically important difference

KEY TAKEAWAYS

The potential impact of efgartigimod PH20 SC treatment or withdrawal of IVIg on deterioration in stage A cannot be conclusively determined due to the ADHERE study design. A study (NCT06637072) investigating transition from IVIg to efgartigimod PH20 SC without disease worsening is ongoing

In all prior treatment groups, the majority of patients improved and responded to efgartigimod in stage A. Though the proportion of participants discontinuing the study/treatment was higher among those who received prior IVIg/SCIg versus those who received prior corticosteroids or were off treatment, the prior IVIg/SCIg subgroup had the highest proportion of aINCAT improvement in stage A with efgartigimod and demonstrated the greatest aINCAT improvement by Week 36 in ADHERE+

In stage A responders, efgartigimod PH20 SC demonstrated aINCAT improvement through Week 36 of ADHERE+ regardless of prior treatment status. Mean change from run-in baseline through ADHERE+ Week 36 improved regardless of prior CIDP treatment (corticosteroids, –1.3; IVIg/SCIg, –1.4; off treatment, –0.9)

PRESENTED AT THE 55TH CONGRESS OF THE ITALIAN SOCIETY OF NEUROLOGY (SIN); OCTOBER 24–28, 2025; PADUA, ITALY

ABBREVIATIONS

aINCAT, adjusted Inflammatory Neuropathy Cause and Treatment; **CI**, confidence interval; **CIDP**, chronic inflammatory demyelinating polyradiculoneuropathy; **ECI**, evidence of clinical improvement; **ECI**, evidence of clinical improvement; **ECMD**, evidence of clinically meaningful deterioration; **EFG**, efgartigimod PH20; **Fc**, fragment crystallizable; **HR**, hazard ratio; **Ig**, immunoglobulin; **IgG**, immunoglobulin G; **INCAT**, Inflammatory Neuropathy Cause and Treatment; **I-RODS**, Inflammatory Rasch-Built Overall Disability Scale; **IVIg**, intravenous immunoglobulin; **MGS**, mean grip strength; **mITT**, modified intention-to-treat; **OLE**, open-label extension; **PBO**, placebo; **PH20**, recombinant human hyaluronidase PH20; **SC**, subcutaneous; **SCIg**, subcutaneous immunoglobulin; **SE**, standard error.

DISCLOSURES AND ACKNOWLEDGMENTS

JAA: Akcea Therapeutics, Alexion, Alnylam Pharmaceuticals, Annexon Biosciences, argenx, CSL Behring, Grifols, Immunovant, ImmuPharma, Johnson & Johnson, Pfizer, Takeda; **JL:** Nothing to declare; **MS:** argenx, Bayer, Biogen Idec, Biotest, CSL Behring, Genzyme, Grifols, Immunovant, Kedrion, Merck, Novartis, Octapharma, PPTA, Roche, Sanofi-Aventis, Teva Pharmaceuticals, UCB; **JTG, GI, ADR, SR, BVH:** Employees of argenx; **SK:** Alexion, argenx, CSL Behring, Takeda; **GL:** Biogen, Chromocell, CSL Behring, Home Biosciences, Janssen, Lilly, Sangamo Therapeutics, Vertex Pharmaceuticals, Zambon; **LQ:** Alnylam Pharmaceuticals, Annexon Biosciences, argenx, Avilar Therapeutics, Biogen, CIBERER, CSL Behring, Dianthus, Fundació La Marató, GBS/CIDP Foundation International, Grifols, Instituto de Salud Carlos III – Ministry of Economy and Innovation (Spain), Janssen, LFB, Lundbeck, Merck, Novartis, Octapharma, Roche, Sanofi, UCB; **NS:** Alnylam Pharmaceuticals, Takeda; **CK:** Alexion, Alnylam Pharmaceuticals, Alpine, Annexon Biosciences, argenx, AstraZeneca, Biogen, Corino Therapeutics, CSL Behring, Genentech, Ionis, Neuroderm, Novo Nordisk, Pfizer, Sanofi, Takeda, UCB, Zai Lab; **TS:** Alexion, Alnylam Pharmaceuticals, argenx, Bayer Vital, Biogen, Bristol Myers Squibb, Celgene, Centogene, CSL Behring, EUROIMMUN, Grifols, Hexal AG, Horizon, Janssen-Cilag, Merck Serono, Novartis, Pfizer, Roche, Sanofi, Siemens, Swedish Orphan Biovitrum, Teva Pharmaceuticals, Viatris; **SR:** Annexon Biosciences, argenx, Beijing Association of Holistic and Integrated Medicine, British Medical Association, CSL Behring, Dianthus Therapeutics, EXCEMED, Fresenius, GBS/CIDP Foundation International, GAIN (Guillain-Barré Syndrome and Related Inflammatory Neuropathies) charity, Hansa Biopharma, Irish Institute of Clinical Neuroscience, Medical Research Council (UK), National Institute of Health Research (NIHR), Pathological Society of Great Britain Ireland, Peripheral Nerve Society, Takeda, UCB, University of Oxford's John Fell Fund, Wellcome Trust; **AE-L:** Alnylam Pharmaceuticals, argenx, CSL Behring, Grifols, LFB, Pfizer, Sanofi; **CH:** argenx, Biogen, Lupin, Roche, UCB; **RY:** Alnylam Japan, CSL Behring, FP Pharm, Japan Tobacco, Kyowa Kirin, Ono Pharmaceutical, Takeda; **PAvD:** Annexon Biosciences, argenx, Grifols, Hansa Biopharma, Octapharma, Prinses Beatrix Spierfonds, Roche, Sanofi, Sanquin, Takeda; **RAL:** Alexion, Annexon Biosciences, argenx, Avilar Therapeutics, BioCryst, Boehringer Ingelheim, CSL Behring, Dianthus Therapeutics, GBS/CIDP Foundation International, Grifols, Immunovant, Intellia Therapeutics, Johnson & Johnson, Medscape, MGFA, Nervosave Therapeutics, Novartis, Nuvig Therapeutics, Sanofi, Seismic Therapeutic, Takeda, TGTX, UpToDate. This trial was sponsored by argenx. Medical writing support was provided by Envision Ignite, an Envision Medical Communications agency, a part of Envision Pharma Group, and was funded by argenx. The authors gratefully acknowledge the trial participants and investigators involved. Previously presented at the Peripheral Nerve Society (PNS) Annual Meeting: May 17–20, 2025, Edinburgh, UK

REFERENCES

1. Rajabally YA. *Immunotargets Ther.* 2024;13:99–110. **2.** Van den Bergh PYK, et al. *Eur J Neurol.* 2010;17:356–63. **3.** Ulrichts P, et al. *J Clin Invest.* 2018;128:4372–86. **4.** Howard JF Jr, et al. *Lancet Neurol.* 2021;20:526–36. **5.** Broome CM, et al. *Lancet.* 2023;402:1648–59. **6.** Allen JA, et al. *Lancet Neurol.* 2024;23:1013–24. **7.** Roopenian DC, Akilesh S. *Nat Rev Immunol.* 2007;7:715–25. **8.** Ward ES, Ober RJ. *Trends Pharmacol Sci.* 2018;39:892–904. **9.** VYVGART HYTRULO. Prescribing information. argenx; 2024. https://argenx.com/content/dam/argenx-corp/products/vyvgart-hytrulo-prescribing-information.pdf. Accessed October 03, 2025.

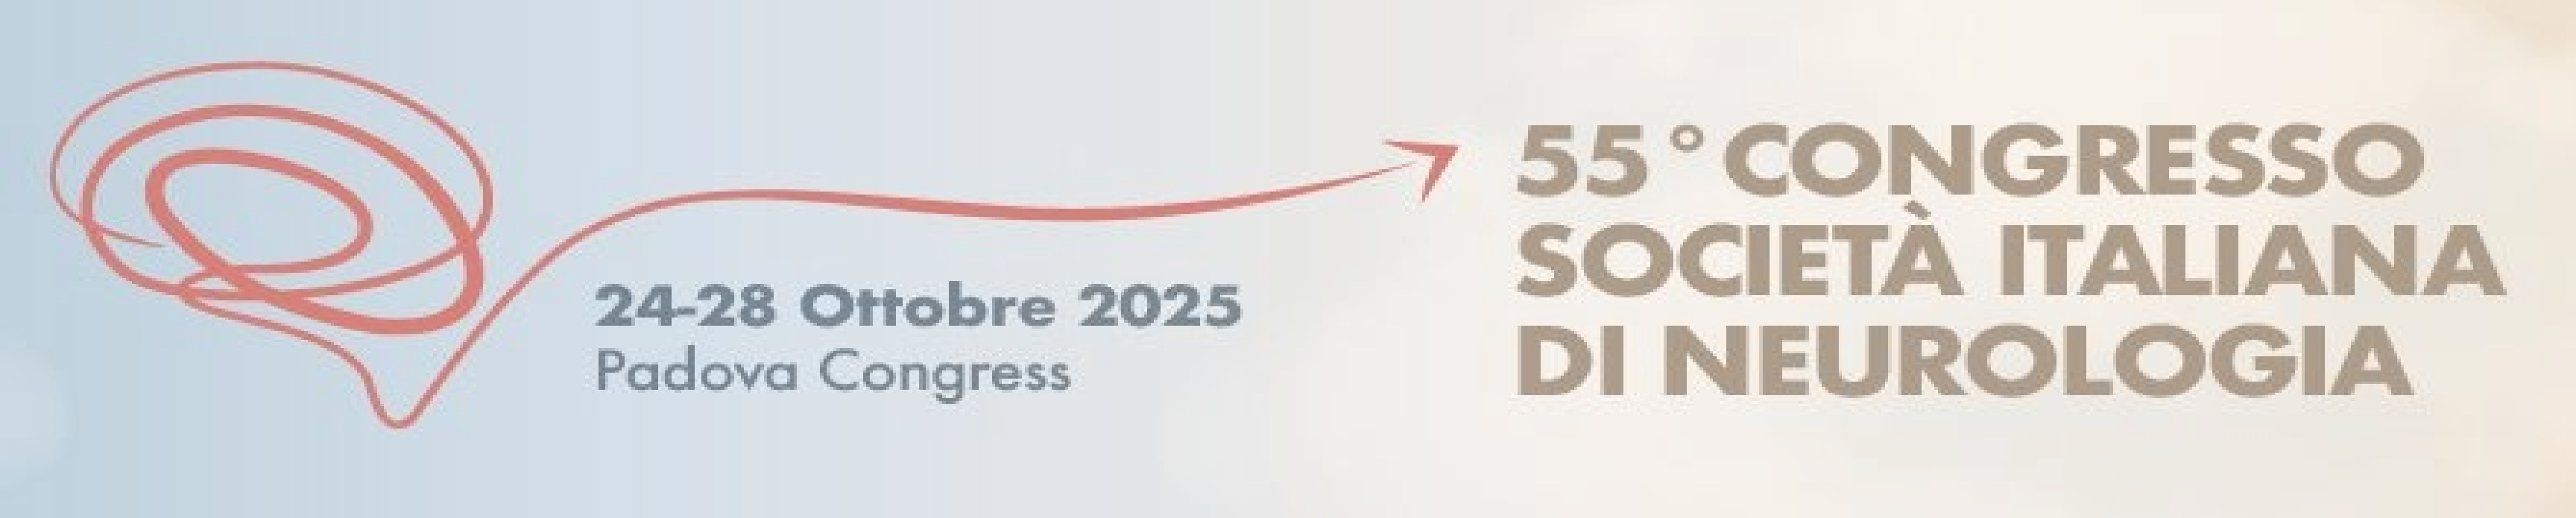

24-28 Ottobre 2025
Padova Congress
55° CONGRESSO SOCIETÀ ITALIANA DI NEUROLOGIA