364

Impact of Atogepant on Migraine Headache Severity for the Preventive Treatment of Episodic Migraine: A Post-Hoc Analysis of the 12-Week ADVANCE and 52-Week Open-Label Trials

Raffaello Masselli* Umer Najib[1], Belinda Savage-Edwards[2], Teshamae Monteith[3], Hsiangkuo Yuan[4], Yingyi Liu[5], Natty Chalermpalanupap[5], Ivonna Reda[5], Brett Dabruzzo[5] and Jessica Ailani[6]

*[1]WVU Headache Center, Morgantown, WV, USA; [2]Rehabilitation and Neurological Services, LLC, Huntsville, AL, USA; [3]Miller School of Medicine, University of Miami, Miami, FL, USA; [4]Jefferson Headache Center, Department of Neurology, Thomas Jefferson University, Philadelphia, PA, USA; [5]AbbVie, North Chicago, IL, USA; *AbbVie srl Rome, Italy. This individual has been added to this publication author by-line, with the permission of the original authors, for the express purpose of conducting the presentation at a local congress or in a local language. He did not contribute to the content of the publication. [6]MedStar Georgetown University Hospital, Washington, DC, USA;*

OBJECTIVE

Evaluate the impact of atogepant 60 mg once daily (QD) on migraine headache day severity in participants with episodic migraine (EM)

CONCLUSIONS

Atogepant treatment reduced the mean number of monthly moderate to severe migraine headache days and mean pain severity of migraine headache days compared with placebo at all timepoints in the 12-week ADVANCE trial

Reductions in the mean number of monthly moderate to severe migraine headache days and mean pain severity of migraine headache days were consistent compared to baseline across 52 weeks in the long-term open-label trial

Atogepant was safe and well tolerated in both trials (previously reported[1,2])

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial. AbbVie funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Brian Neel, PhD, of AbbVie. Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: UN is a consultant for and received honoraria from AbbVie and Pfizer. B S-E has received research grant support from AbbVie, Amgen, Eli Lilly, IQVIA, and the Vector Psychometric Group. She presently is on the Speaker Bureau for Eisai and Eli Lilly and has been on the Speaker Bureau in the past for Biohaven, Impel and AbbVie. She is on the Advisory Board for Eli Lilly and is a Consultant for AbbVie and Impel. She has stock options for Eli Lilly and AbbVie and is compensated for serving as an Associate Editor for Neurology Clinical Practice. HY in the past 24 months, received grants from NIH (R44NS115460), American Headache Society, and royalties from Cambridge University Press, and Medlink. He served as site investigator for Teva, AbbVie, Ipsen, Parema, and consultant for Salvia, Pfizer, AbbVie, and Cerenovous. TM has indirect funding to institution for involvement in industry research (site PI) for AbbVie, Eli Lily, Ipsen, and Rehaler; she has educational grants from AbbVie and Amgen. She is on the board of directors for the Florida Society of Neurology and was previously on the International Headache Society board of directors. She is on the editorial board for the American Migraine Foundation, Brain and Life Magazine, and Neurology. She is an associate editor for Cephalalgia, Confinia Cephalalgica, and Continuum Audio. She is a deputy editor for Neurology Minute podcast. She has done advisory work for AbbVie, eNeura/Aruene, Merz, Pfizer, and Teva. Other compensated speaking activities are for the American Academy of Neurology, American Headache Society, and Medscape. JA has received clinical trial grants as a principal investigator from Allergan/AbbVie, American Migraine Foundation, Biohaven, Eli Lilly, Satsuma, and Zosano; has received stock options for consulting from CtrlM; and has received honoraria from the following: Alder/Lundbeck (speaking, consulting), Allergan/AbbVie (speaking, consulting), Amgen (speaking, consulting), Biohaven (speaking, consulting), GlaxoSmithKline (consulting), Eli Lilly and company (speaking, consulting), Impel (consulting), Theranica (consulting), Axsome (consulting), Neso (consulting), Aeon (data advisory board), Teva (speaking, consulting), Satsuma (consulting), Current Pain and Headache Reports (section editor), NeurologyLive (editorial board), SELF (medical advisory board), Medscape (advisory board). YL, NC, BD, and IR are employees of AbbVie and may hold AbbVie stock.
These data were previously presented at the American Academy of Neurology Annual Meeting (AAN), April 5-9, 2025, San Diego, CA, USA. (REF DV-014609)

INTRODUCTION

- Atogepant is an oral, calcitonin gene-related peptide (CGRP) receptor antagonist approved for the preventive treatment of migraine[3,4]
- ADVANCE was a phase 3, randomized, double-blind, placebo-controlled, 12-week trial evaluating atogepant 60 mg QD for the preventive treatment of EM[1]
- The long-term trial was a phase 3, randomized, 52-week, open-label trial evaluating atogepant 60 mg QD for the preventive treatment of EM[2]
- This post-hoc analysis used clinical trial data from both trials to evaluate the impact of atogepant on migraine headache day severity

1. Ailani J et al. N Engl J Med. 2021;385(8):695-706
2. Ashina M et al. Headache. 2023;63:79-88
3. QULIPTA (atogepant). Package Insert. AbbVie Inc.; 2023
4. AQUIPTA (atogepant). Summary of Product Characteristics. AbbVie Inc.; 2024

METHODS

Study Design and Participants

- This post hoc analysis included participants with EM from the 12-week ADVANCE trial and 52-week, open-label trial
 - Outcomes are presented for atogepant 60 mg QD dose and placebo (in ADVANCE)
- The modified intent-to-treat (mITT) population consisted of all randomized participants who received ≥1 dose of study intervention, had an evaluable baseline period of eDiary data, and had ≥1 evaluable post-baseline 4-week period of eDiary data during the double-blind or open-label treatment periods

Outcomes

- Change from baseline in the mean number of monthly moderate to severe migraine headache days
 - Migraine headache pain severity was defined as the worst pain intensity participants reported on any headache day with a scale of 1 (mild), 2 (moderate), and 3 (severe)
- Change from baseline in mean pain severity of migraine headache days
 - If the number of monthly migraine headache days at any post-baseline period were reduced from baseline, a pain severity score of 0 was imputed for the resolved migraine headache days

Statistical Analyses

- Atogepant vs placebo comparisons were analyzed using a mixed-effects model for repeated measures (MMRM) for change from baseline. The model includes baseline monthly moderate/severe migraine days as a covariate, prior exposure to migraine prevention medications (Y/N), treatment group and visit (month) as fixed factors, and treatment group-by-visit and baseline-by-visit as interaction terms, with an unstructured covariance matrix. *P*-values are from the test between the atogepant dose group and placebo

RESULTS

Moderate to Severe Migraine Headache Days (mITT Population)

Figure 1: Mean Number of Monthly Moderate to Severe Migraine Headache Days at Baseline and Weeks 1-4, 5-8, and 9-12 (ADVANCE)

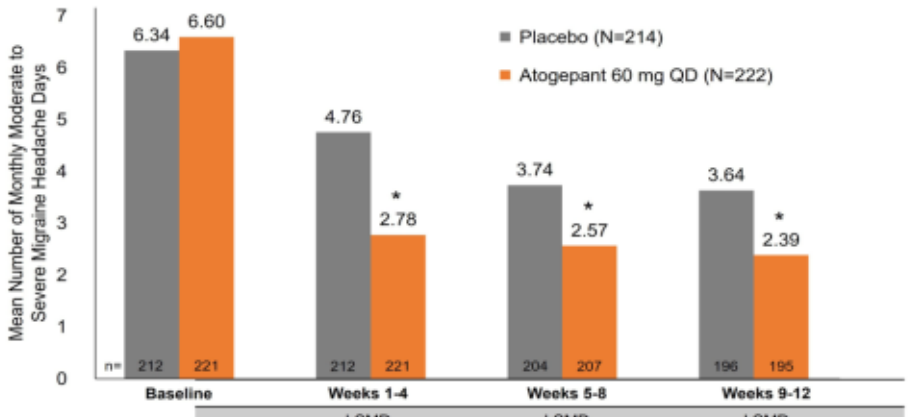
Mean Number of Monthly Moderate to Severe Migraine Headache Days
Placebo (N=214)
Atogepant 60 mg QD (N=222)
6.34
6.60
4.76
2.78
3.74
2.57
3.64
2.39
n= 212 221
212 221
204 207
196 195
Baseline
Weeks 1-4
Weeks 5-8
Weeks 9-12

Atogepant 60 mg QD vs placebo	LSMD (95% CI)	*P*-value	LSMD (95% CI)	*P*-value	LSMD (95% CI)	*P*-value
	-2.08 (-2.64, -1.52)	<0.0001	-1.21 (-1.77, -0.66)	<0.0001	-1.20 (-1.78, -0.63)	<0.0001

CI, confidence interval; LSMD, least squares mean difference; QD, once daily.

- At Weeks 9–12 in ADVANCE, the number of monthly moderate to severe migraine headache days was reduced by 64% with atogepant treatment compared to baseline (vs. 43% reduction with placebo)

Figure 2: Mean Number of Monthly Moderate to Severe Migraine Headache Days at Baseline and 4-Week Intervals (52-Week Open-Label Trial)

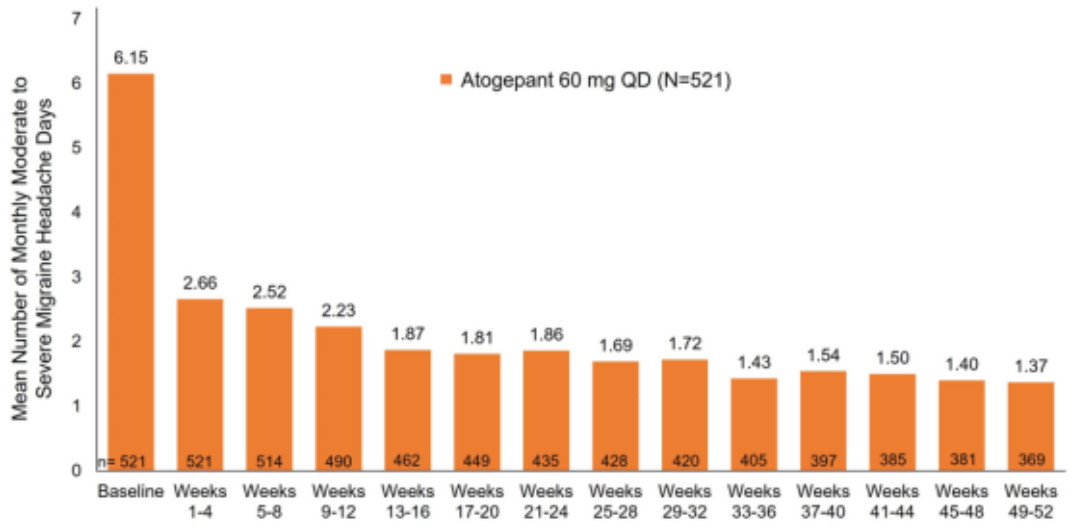
Mean Number of Monthly Moderate to Severe Migraine Headache Days
Atogepant 60 mg QD (N=521)
6.15 2.66 2.52 2.23 1.87 1.81 1.86 1.69 1.72 1.43 1.54 1.50 1.40 1.37
n= 521 521 514 490 462 449 435 428 420 405 397 385 381 369
Baseline
Weeks 1-4
Weeks 5-8
Weeks 9-12
Weeks 13-16
Weeks 17-20
Weeks 21-24
Weeks 25-28
Weeks 29-32
Weeks 33-36
Weeks 37-40
Weeks 41-44
Weeks 45-48
Weeks 49-52

CI, confidence interval; LSMD, least squares mean difference; QD, once daily.

- In the open-label trial, the mean number of monthly moderate to severe migraine headache days was reduced by 57% at Weeks 1–4 compared to baseline, which improved to a 78% reduction at Weeks 49–52

Mean Pain Severity of Migraine Headache Days (mITT Population)

Figure 3: Change from Baseline in Mean Pain Severity of Migraine Headache Days, Including Resolved Migraine Days, at Weeks 1-4, 5-8, and 9-12 (ADVANCE)

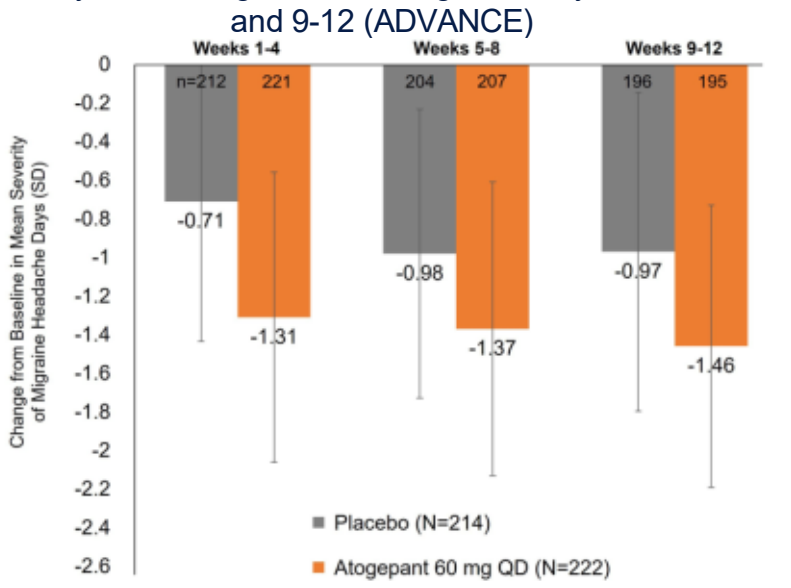
Weeks 1-4
Weeks 5-8
Weeks 9-12
Change from Baseline in Mean Severity of Migraine Headache Days (SD)
n=212 221
204 207
196 195
-0.71
-1.31
-0.98
-1.37
-0.97
-1.46
Placebo (N=214)
Atogepant 60 mg QD (N=222)

SD, standard deviation; QD, once daily.

- Mean baseline (SD) in pain severity for placebo was 2.14 (0.40) and for atogepant, 2.15 (0.40)
- The change from baseline in mean pain severity of migraine headache days, including resolved days, was-1.46 for atogepant and -0.97 for placebo at Weeks 9-12

Figure 4: Change from Baseline in Mean Pain Severity of Migraine Headache Days, Including Resolved Migraine Days, at 4-Week Intervals (52-Week Open-Label Trial)

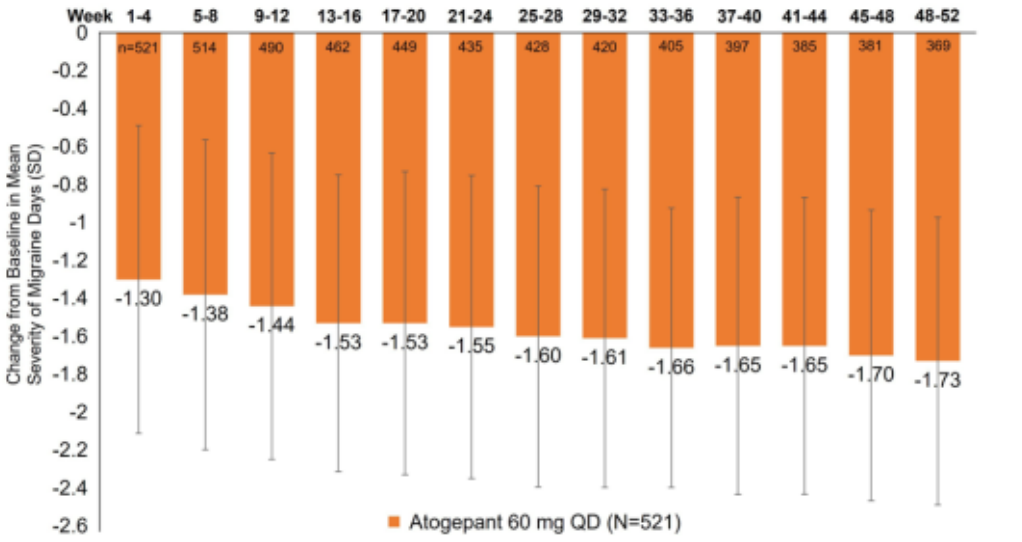
Week 1-4 5-8 9-12 13-16 17-20 21-24 25-28 29-32 33-36 37-40 41-44 45-48 48-52
n=521 514 490 462 449 435 428 420 405 397 385 381 369
-1.30 -1.38 -1.44 -1.53 -1.53 -1.55 -1.60 -1.61 -1.66 -1.65 -1.65 -1.70 -1.73
Change from Baseline in Mean Severity of Migraine Days (SD)
Atogepant 60 mg QD (N=521)

SD, standard deviation; QD, once daily.

- Mean baseline (SD) in pain severity was 2.17 (0.43)
- The change from baseline in mean pain severity of migraine headache days, including resolved days, was 1.30 at Weeks 1–4 and decreased to -1.73 at Weeks 49–52

Presented at the Italian Society of Neurology (SIN) – 55° National Congress, October, 24-25, 2025- Padova, Italy. (CLR-004513)

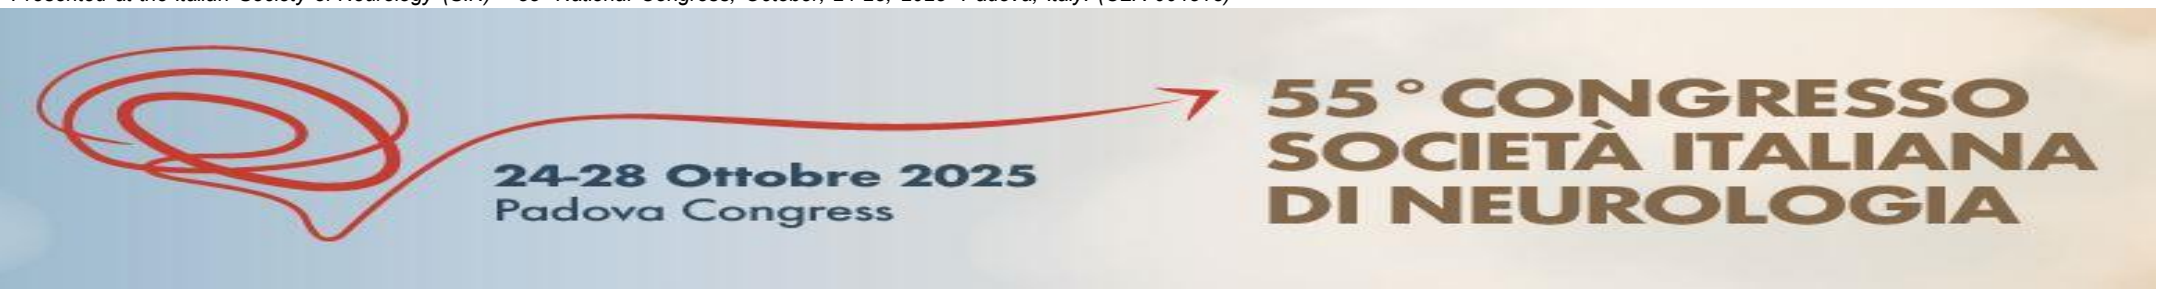
24-28 Ottobre 2025
Padova Congress
55° CONGRESSO
SOCIETÀ ITALIANA
DI NEUROLOGIA