Poster **457**

Quality of life in patients with generalised myasthenia gravis receiving rozanolixizumab: *Post hoc* analysis of MycarinG

Laura Fionda[1], Carlo Antozzi[2], Elena Cortés-Vicente[3], Robert M. Pascuzzi[4], Sabrina Sacconi[5], Kimiaki Utsugisawa[6], John Vissing[7], Tuan Vu[8], Jos Bloemers[9], Fiona Grimson[10], Thaïs Tarancón[11] and Vera Bril[12]

[1]Neuromuscular and Rare Disease Center, Neurology Unit, AOU Sant'Andrea, Sapienza University, Rome Italy; [2]Immunotherapy and Apheresis Unit and Neuroimmunology and Muscle Pathology Unit, Fondazione Istituto di Ricovero e Cura a Carattere Scientifico, Istituto Neurologico Carlo Besta, Milan, Italy; [3]Neuromuscular Diseases Unit, Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; [4]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [5]Université Côte d'Azur, Peripheral Nervous System & Muscle Department, Pasteur 2 Hospital, Centre Hospitalier Universitaire de Nice, Nice, France; [6]Department of Neurology, Hanamaki General Hospital, Hanamaki, Japan; [7]Copenhagen Neuromuscular Center, Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [8]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [9]UCB, Brussels, Belgium; [10]UCB, Slough, UK; [11]UCB, Madrid, Spain; [12]Ellen and Martin Prosserman Centre for Neuromuscular Diseases, Toronto General Hospital, University of Toronto, Toronto, Ontario, Canada

SIN 2025, Padua, Italy; 24–28 October 2025

Introduction

- MG is a chronic autoimmune disease characterised by fluctuating muscle weakness fatigability that can significantly impact many aspects of patients' HRQoL[1,2]
- The MG-QoL15r is a PRO instrument designed to investigate the physiological, social and emotional impact of MG-specific impairment, comprising 15 items (total score: 0–30), where higher scores reflect worse HRQoL[3]
- In the MycarinG study (NCT03971422), rozanolixizumab significantly improved MG-specific outcomes versus placebo in patients with gMG[4]
- Here, we evaluate the impact of rozanolixizumab on different facets of HRQoL in patients with gMG, using three themes of the MG-QoL15r in a *post hoc* analysis of MycarinG data

Methods

- Patients were randomised 1:1:1 to receive once-weekly SC rozanolixizumab 7 mg/kg, rozanolixizumab 10 mg/kg or placebo for 6 weeks (to Day 43), followed by an 8-week observation period
- LS mean (95% CI) CFB in MG-QoL15r at Day 43 was determined *post hoc* for the three treatment groups using an MMRM ANCOVA model
- The 15 items of the MG-QoL15r were grouped into three themes: physical (score range: 0–14), social (score range: 0–8) and emotional (score range: 0–8; **Table 1**)
 - Descriptive analyses of the CFB in MG-QoL15r scores at Day 43 for each theme were conducted

Results

- Overall, 200 patients received rozanolixizumab 7 mg/kg (n=66), rozanolixizumab 10 mg/kg (n=67) or placebo (n=67)
- LS mean (SE) CFB in MG-QoL15r total score at Day 43 was greater in both rozanolixizumab groups than in the placebo group (**Figure 1**)
- Across the three themes, mean CFB in scores were greater in the rozanolixizumab groups than in the placebo group (**Figure 2**)
- At baseline, the majority of patients reported MG symptoms impacting their QoL across all three themes represented by scores ≥1 (rozanolixizumab 7 mg/kg: 90.9% [n=60]; rozanolixizumab 10 mg/kg: 90.9% [n=60]; placebo: 94.0% [n=63])
- At Day 43, the proportions of patients who reported no impact of symptoms on QoL across each of the three themes were greater for both rozanolixizumab dose groups compared with the placebo group (**Table 2**)
 - A complete absence of HRQoL impairment, indicated by scores of 0 across all three themes, was reported at Day 43 in 7.7% (n=5) and 10.6% (n=7) of patients in the rozanolixizumab 7 mg/kg and 10 mg/kg groups, respectively, compared with no patients in the placebo group
- Overall, rozanolixizumab was generally well tolerated and demonstrated an acceptable safety profile
 - TEAEs occurred in 81.3% (n=52/64), 82.6% (n=57/69) and 67.2% (n=45/67) of patients treated with rozanolixizumab 7 mg/kg, 10 mg/kg and placebo, respectively; most were mild or moderate
 - The most frequent TEAEs were headache (rozanolixizumab 7 mg/kg: 45.3%; 10 mg/kg: 37.7%; placebo: 19.4%), diarrhoea (7 mg/kg: 25.0%; 10 mg/kg: 15.9%; placebo: 13.4%) and pyrexia (7 mg/kg: 12.5%; 10 mg/kg: 20.3%; placebo: 1.5%)

Summary and conclusions

- Improvements in MG-QoL15r total score were greater and nominally statistically significant in the rozanolixizumab treatment groups compared with the placebo group
- Mean CFB at Day 43 was greater in physical, social and emotional themes of the MG-QoL15r for both rozanolixizumab groups compared with placebo
- Across the three themes, greater proportions of patients in the rozanolixizumab groups achieved an absence of impact of symptoms on HRQoL as measured by the MG-QoL15r
- These data demonstrate that patients benefit from rozanolixizumab treatment across the breadth of impacts that MG symptoms have on patients' HRQoL

Figure 1 LS mean CFB in MG-QoL15r total score at Day 43 was greater with both rozanolixizumab doses than with placebo

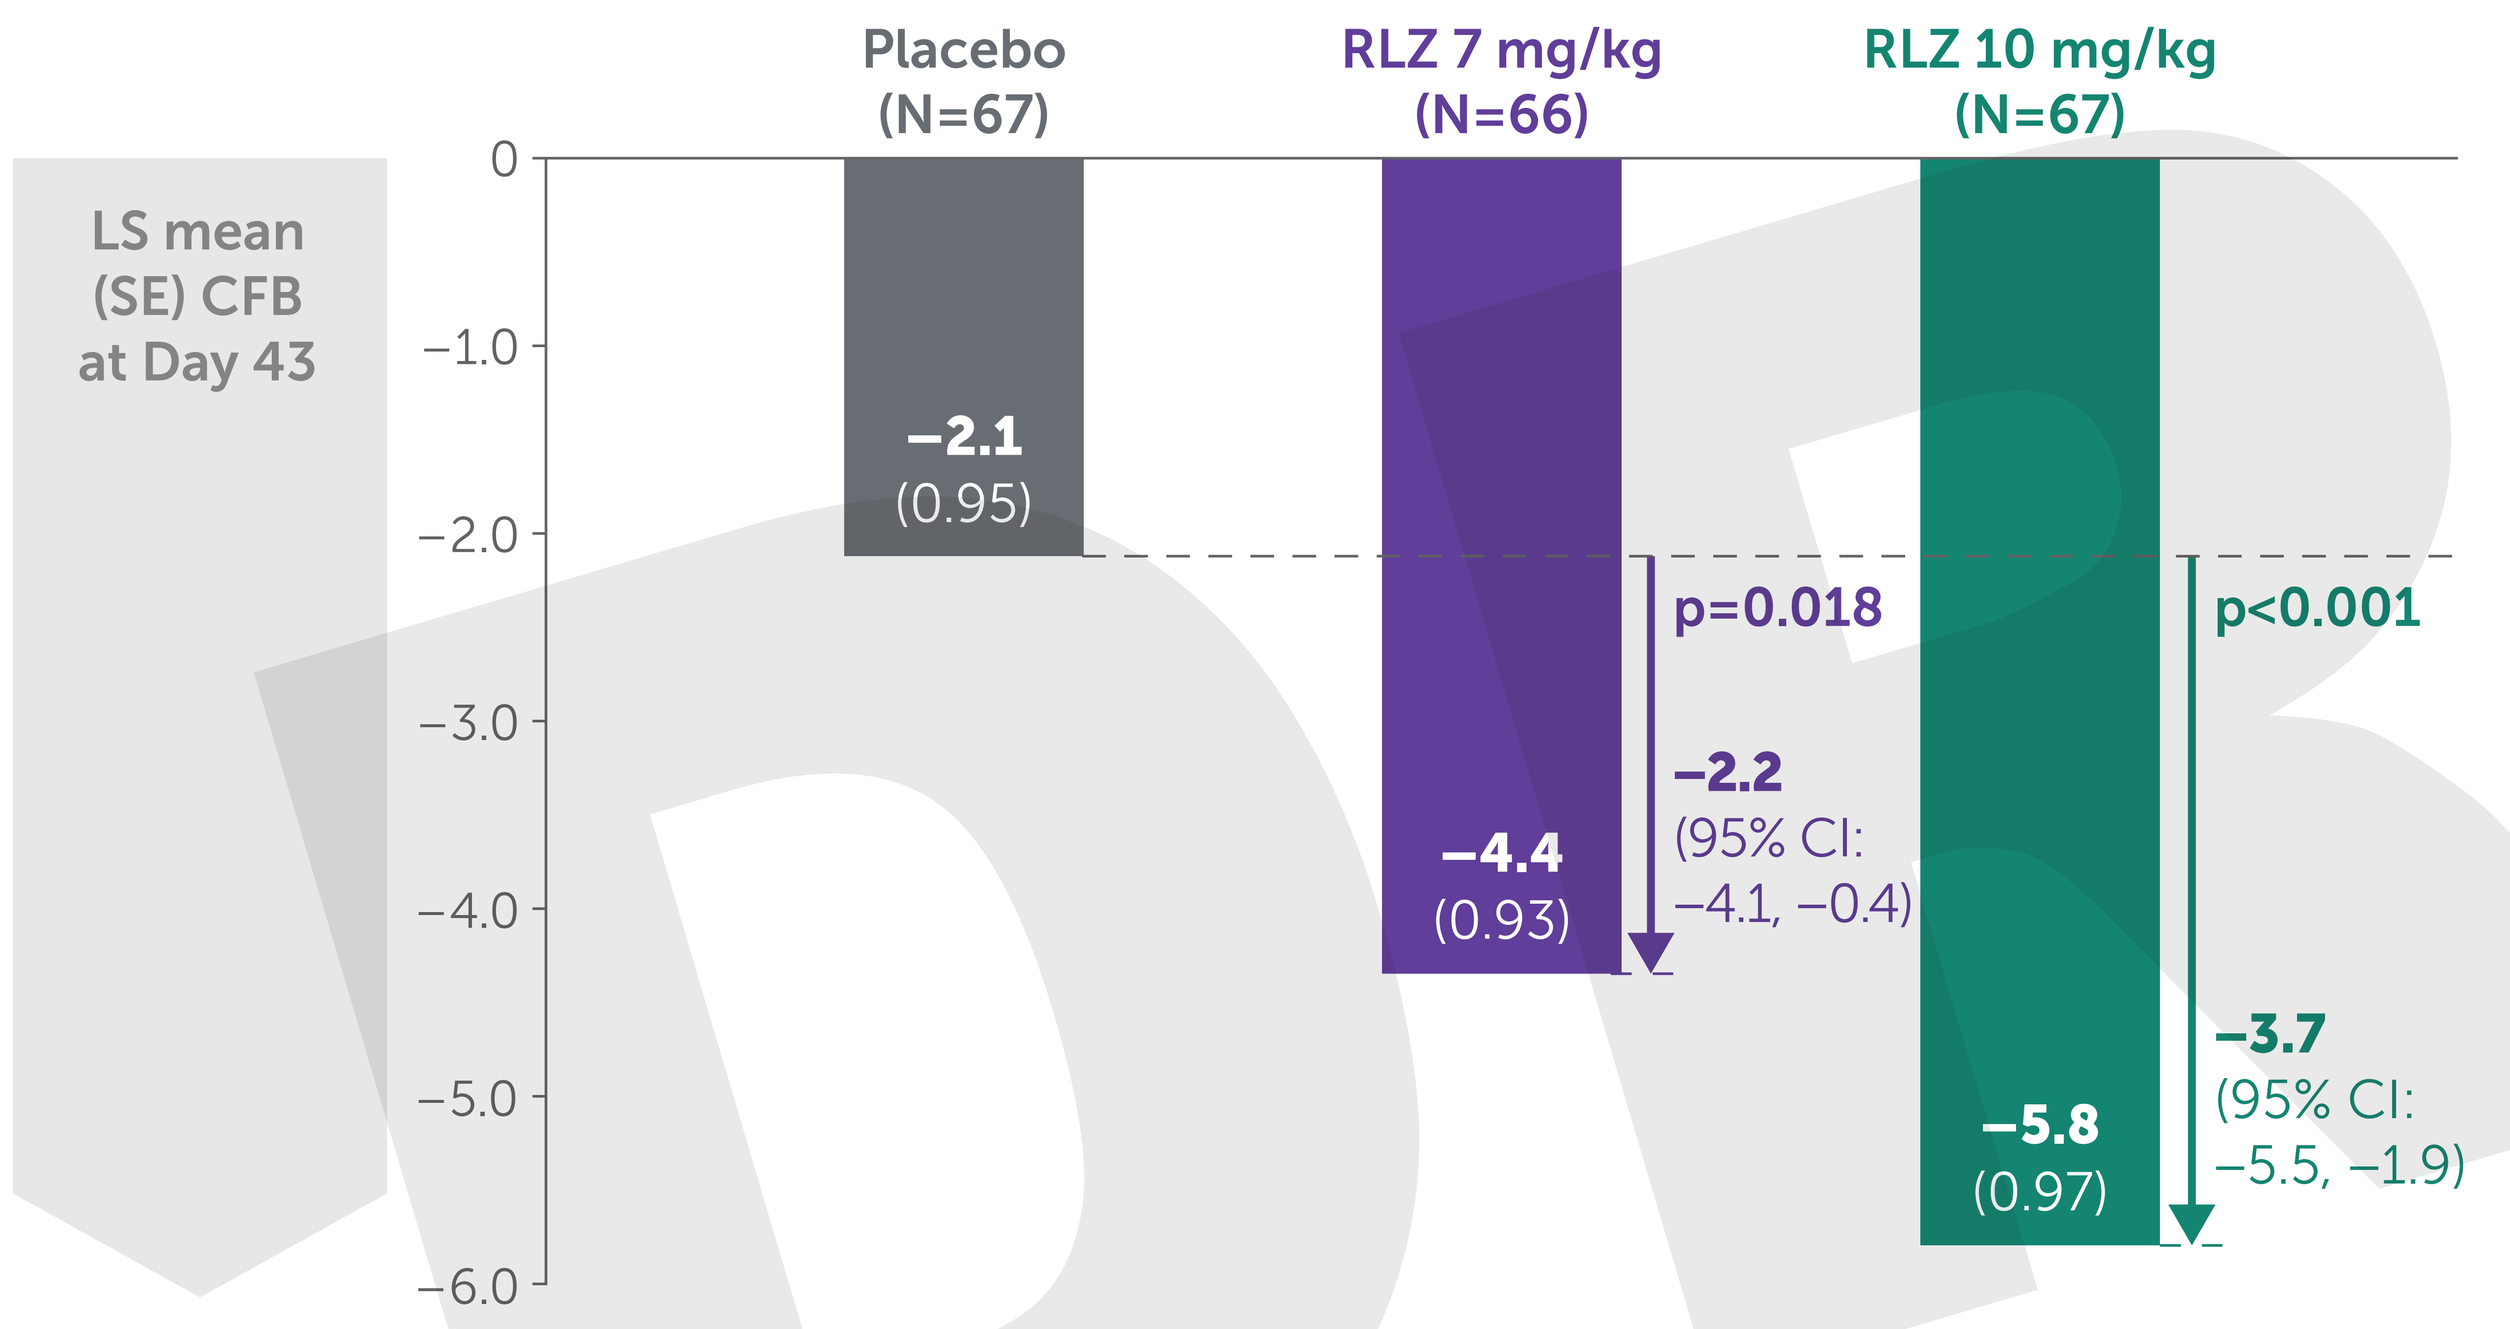
Placebo
(N=67)
RLZ 7 mg/kg
(N=66)
RLZ 10 mg/kg
(N=67)
LS mean
(SE) CFB
at Day 43
0
−1.0
−2.0
−3.0
−4.0
−5.0
−6.0
−2.1
(0.95)
−4.4
(0.93)
−5.8
(0.97)
p=0.018
−2.2
(95% CI:
−4.1, −0.4)
p<0.001
−3.7
(95% CI:
−5.5, −1.9)

	Placebo (n=67)	RLZ 7 mg/kg (n=66)	RLZ 10 mg/kg (n=66)
Baseline total score, mean (SD)	**15.0** (6.4)	**15.7** (7.7)	**15.5** (6.6)

Baseline MG-QoL15r data were missing for one patient in the RLZ 10 mg/kg dose group.

Figure 2 Across the three themes of the MG-QoL15r, mean CFB at Day 43 was greater with both rozanolixizumab doses than with placebo

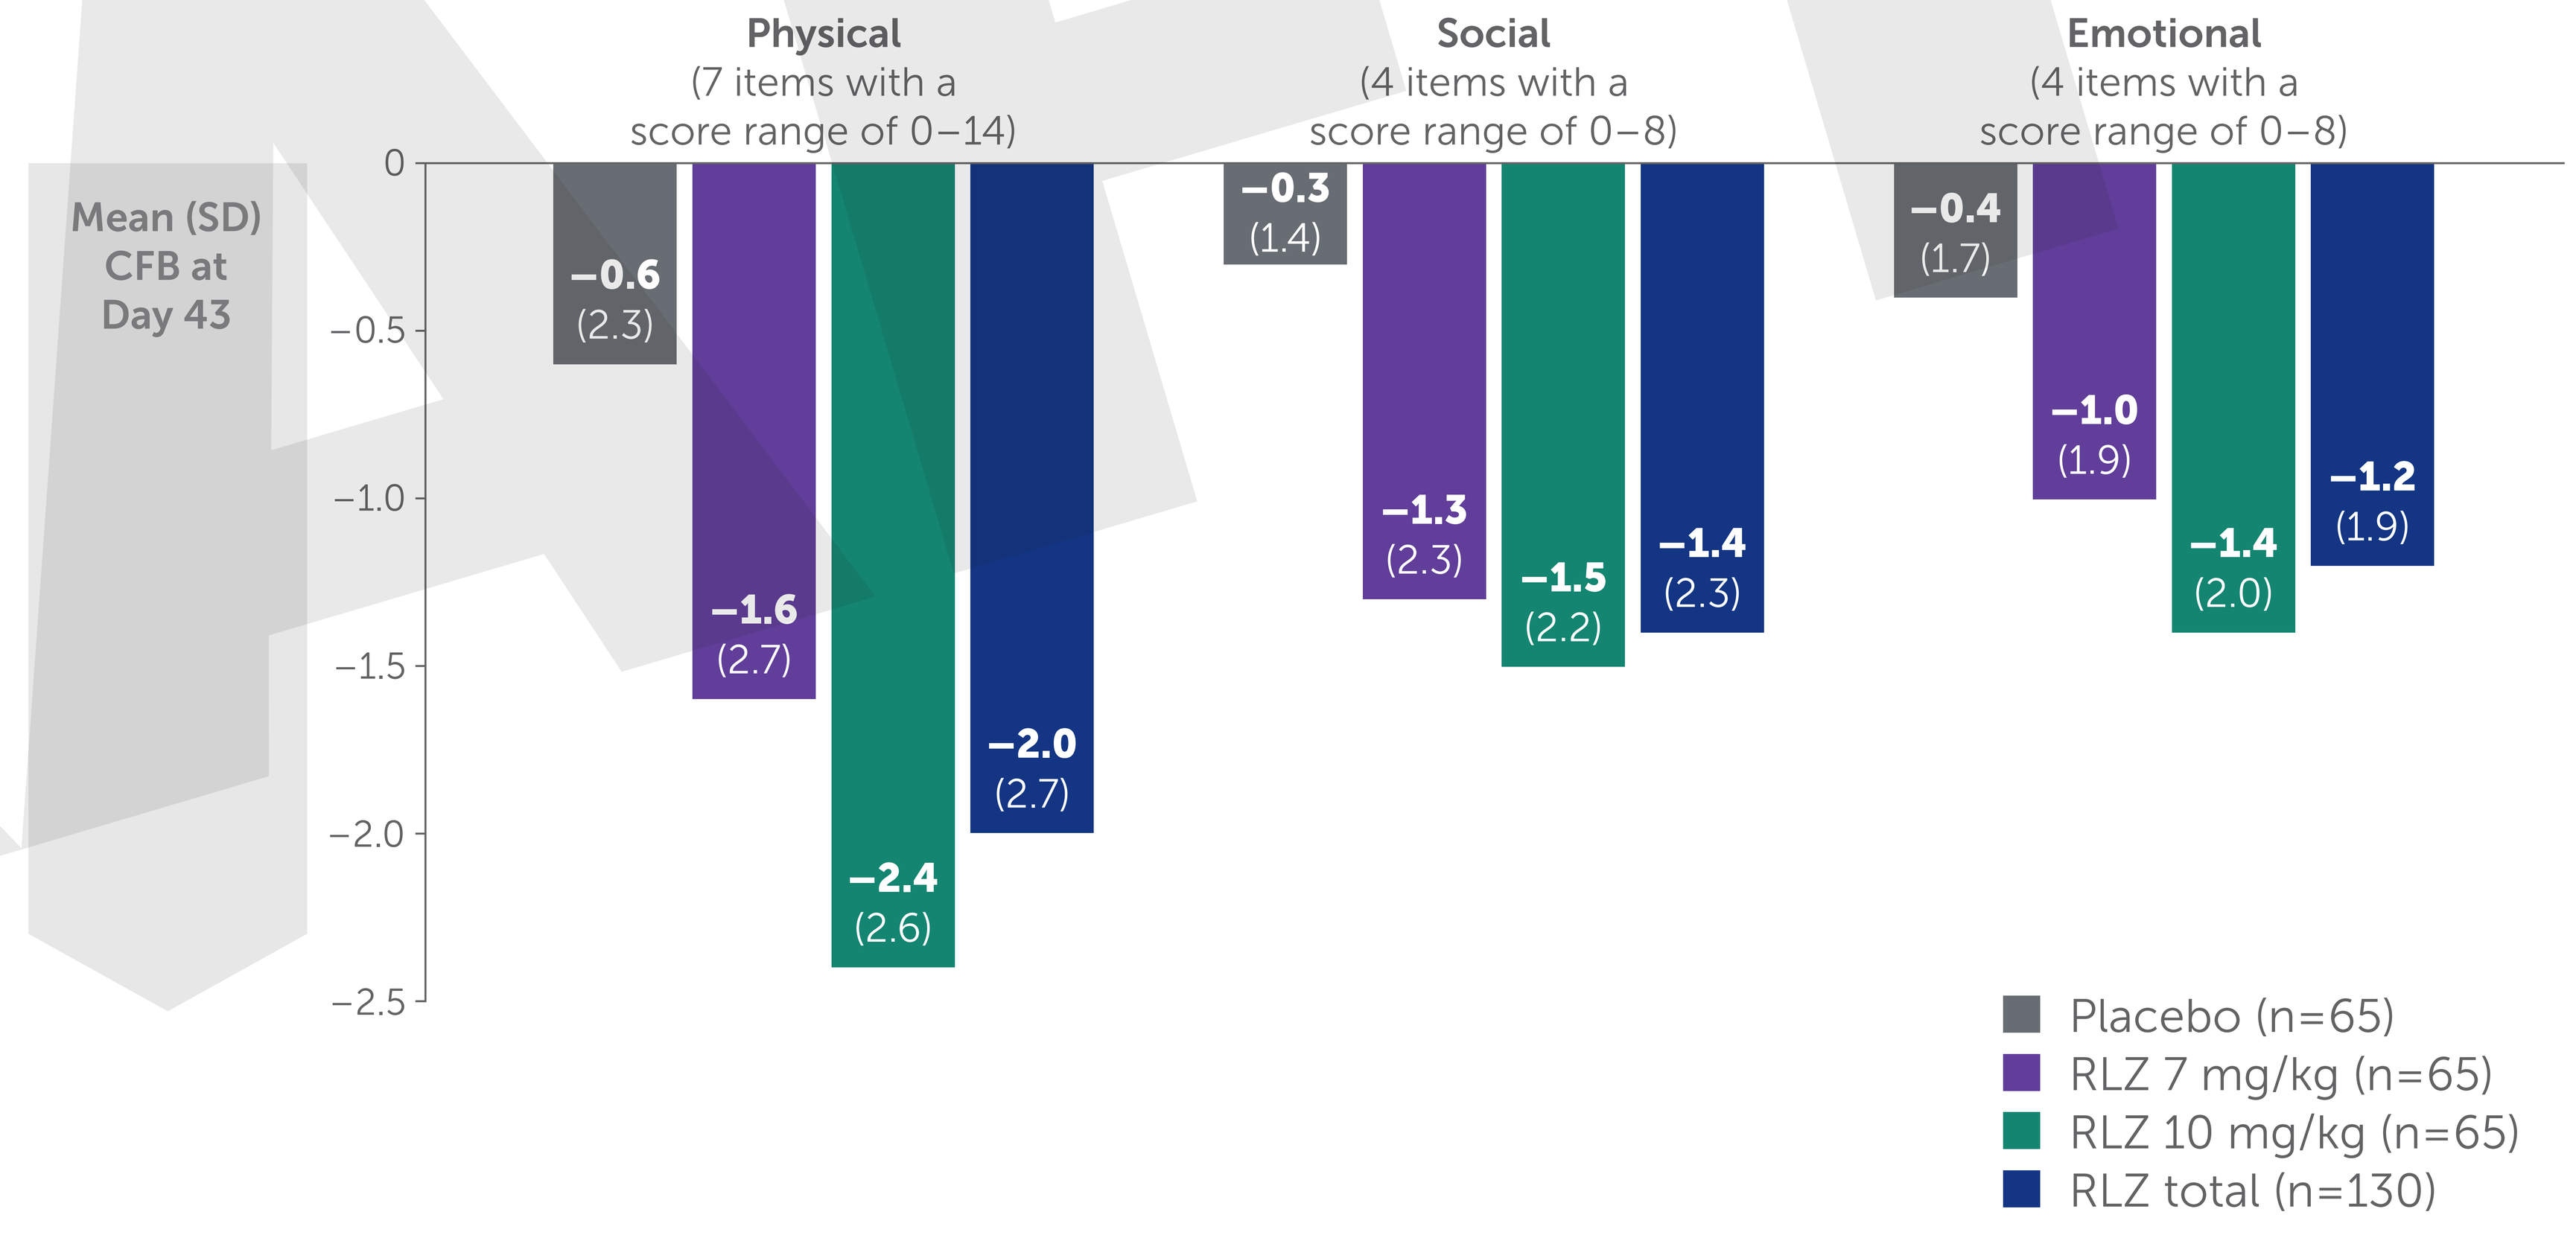
Physical
(7 items with a
score range of 0–14)
Social
(4 items with a
score range of 0–8)
Emotional
(4 items with a
score range of 0–8)
Mean (SD)
CFB at
Day 43
0
−0.5
−1.0
−1.5
−2.0
−2.5
−0.6
(2.3)
−1.6
(2.7)
−2.4
(2.6)
−2.0
(2.7)
−0.3
(1.4)
−1.3
(2.3)
−1.5
(2.2)
−1.4
(2.3)
−0.4
(1.7)
−1.0
(1.9)
−1.4
(2.0)
−1.2
(1.9)
Placebo (n=65)
RLZ 7 mg/kg (n=65)
RLZ 10 mg/kg (n=65)
RLZ total (n=130)

Table 1 MG-QoL15r items were assigned to three themes

Please indicate how true each statement has been (over the past few weeks)	Physical	Social	Emotional
1. I am frustrated by my MG			X
2. I have trouble using my eyes because of my MG (e.g. double vision)	X		
3. I have trouble eating because of my MG	X		
4. I have limited my social activity because of my MG		X	
5. My MG limits my ability to enjoy hobbies and fun activities		X	
6. I have trouble meeting the needs of my family because of my MG		X	
7. I have to make plans around my MG		X	
8. I am bothered by limitations in my performing at work (include work at home) because of my MG			X
9. I have difficulty speaking due to MG	X		
10. I have lost some personal independence because of my MG (e.g. driving, shopping, running errands)	X		
11. I am depressed about my MG			X
12. I have trouble walking due to MG	X		
13. I have trouble getting around in public places because of my MG	X		
14. I feel overwhelmed by my MG			X
15. I have trouble performing my personal grooming needs due to MG	X		

Table 2 Across the three themes, there was a greater increase in the proportion of patients with no impact of symptoms on QoL (score of 0) at Day 43 in both rozanolixizumab dose groups than with placebo

0–<5% | 5–<10% | 10–<15% | 15–<20% | ≥20%

	Patients with score of 0 at baseline, n (%)			Patients with score of 0 at Day 43, n (%)		
Theme	Placebo	RLZ 7 mg/kg	RLZ 10 mg/kg	Placebo	RLZ 7 mg/kg	RLZ 10 mg/kg
Physical	**0** (0)	**4** (6.1)	**2** (3.0)	**2** (3.1)	**11** (16.9)	**12** (18.2)
Social	**3** (4.5)	**3** (4.5)	**3** (4.5)	**3** (4.6)	**11** (16.9)	**14** (21.2)
Emotional	**1** (1.5)	**2** (3.0)	**4** (6.1)	**2** (3.1)	**12** (18.5)	**15** (22.7)
Physical and social	**0** (0)	**2** (3.0)	**1** (1.5)	**0** (0)	**8** (12.1)	**9** (13.4)
Physical and emotional	**0** (0)	**1** (1.5)	**0** (0)	**0** (0)	**6** (9.1)	**9** (13.4)
Social and emotional	**0** (0)	**1** (1.5)	**2** (3.0)	**0** (0)	**7** (10.6)	**10** (14.9)
Physical, social and emotional	**0** (0)	**1** (1.5)	**0** (0)	**0** (0)	**5** (7.7)	**7** (10.6)

Abbreviations: ANCOVA, analysis of covariance; CFB, change from baseline; CI, confidence interval; gMG, generalised myasthenia gravis; HRQoL, health-related quality of life; LS, least squares; MG, myasthenia gravis; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item Scale – Revised; MMRM, mixed model for repeated measures; PRO, patient-reported outcome; SC, subcutaneous; SD, standard deviation; SE, standard error.

Acknowledgements: This study was funded by UCB. The authors acknowledge Annabel Dimmock, MBiol, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB, Slough, UK, for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Laura Fionda has received funding for travel and meeting attendance or advisory board participation from Alexion Pharmaceuticals, argenx and Dianthus. Carlo Antozzi has received funding for congress and Institutional Review Board participation from Alexion Pharmaceuticals, argenx, Biogen, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson) and UCB. Elena Cortés Vicente receives public speaking honoraria and compensation for advisory boards and/or consultation fees from Alexion Pharmaceuticals, argenx, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and UCB. Robert M. Pascuzzi is Professor Emeritus of Neurology at Indiana University and receives compensation for his professional work from Indiana University Health. He has no financial relationship with any pharmaceutical company and receives no compensation from any pharmaceutical company (present or past). Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organisation Medical Education Resources (an educational organisation with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation, or publication. Sabrina Sacconi has nothing to disclose. Kimiaki Utsugisawa has served as a paid Consultant for argenx, Chugai Pharmaceutical, HanAll Biopharma, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck, Mitsubishi Tanabe Pharma, UCB and Viela Bio (now Amgen); he has received speaker honoraria from Alexion Pharmaceuticals, argenx, the Japan Blood Products Organization and UCB. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research and travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Atamyo Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals and UCB, and has served as a speaker for Alexion/AstraZeneca Rare Disease, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics and ImmunAbs. Jos Bloemers, Fiona Grimson and Thaïs Tarancón are employees and shareholders of UCB. Vera Bril is a Consultant for Akcea, Alexion Pharmaceuticals, Alnylam, argenx, CSL, Grifols, Immunovant, Ionis, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson), Novo Nordisk, Octapharma, Pfizer, Powell Mansfield, Roche, Sanofi, Takeda Pharmaceuticals and UCB. She has received research support from Akcea, Alexion Pharmaceuticals, argenx, CSL, Grifols, Immunovant, Ionis, Momenta (now Johnson & Johnson), Octapharma, Takeda Pharmaceuticals, UCB and Viela Bio (now Amgen).

References: 1. Gilhus NE, et al. Nat Rev Dis Primers. 2019;5(1):30. 2. Jackson K, et al. Neurol Ther. 2023;12:107–128. 3. Burns TM, et al. Muscle Nerve. 2016;54:1015–1022. 4. Bril V, et al. Lancet Neurol. 2023;22(5):383–394.

Please use this QR code to download a PDF of the poster.

These data were previously presented at EAN 2025, Helsinki, Finland; 21–24 June 2025