752

Pain Relief in Patients < 55 and ≥ 55 Years With Spasticity Treated With OnabotulinumtoxinA: Results From the ASPIRE Study

Manuela Gorini*; Jörg Wissel[1]; Esther Duarte[2]; Wuwei Feng[3]; Marc Schwartz[4]; Mariana Nelson[5]; Tiziana Musacchio[6]; Cassandra List[7]

*[1]Neurology and Psychosomatic at Wittenbergplatz, Ber[1]lin and University of Potsdam, Germany; [2]Department of Physical Medicine and Rehabilitation, Hospitals del Mar i l'Esperança, Barcelona, Spain; [3]Department of Neurology, Duke University School of Medicine, Durham, NC, USA; [4]MS Biostatistics, LLC, Clermont, FL, USA; [5]AbbVie Inc., Irvine, CA, USA; [6]AbbVie Inc., Rome, Italy; *AbbVie srl Rome, Italy. This individual has been added to this publication author by-line, with the permission of the original authors, for the express purpose of conducting the presentation at a local congress or in a local language. She did not contribute to the content of the publication.[7]Brooks Rehabilitations, Jacksonville, FL, USA*

OBJECTIVE

To assess the effect of onabotulinumtoxinA (onabotA) on pain relief in spasticity patients stratified by their median age <55 years and ≥55 years from the ASPIRE study

CONCLUSIONS

In this subanalysis of the ASPIRE study, onabotA treatment reduced pain in spasticity patients < 55 years and ≥ 55 years who had pain at baseline as measured by the NPRS and DAS pain subscale

Overall, most patients and physicians in both age groups were satisfied/extremely satisfied that onabotA helped the pain and would continue to use it to treat spasticity

This subanalysis of ASPIRE provides valuable real-world evidence on onabotA utilization and effectiveness on pain relief in spasticity patients regardless of age groups (< 55 years and ≥ 55 years), with no new safety signals observed

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial. AbbVie funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Gina E. Elsen, PhD, of AbbVie. Editorial assistance was provided by Angel T. Hadsell, of AbbVie. Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: **JW** has received honoraria for being on the speaker's bureaus and advisory boards for AbbVie, Ipsen, Medtronic, and Merz; **ED** has received fees for study participation from Allergan, Dysport, and Merz; **WF** has received research grants from the National Institutes of Health (NIH) and AHA; **MS** is a biostatistical consultant for AbbVie; **MN, TM and MG** are full-time employees of AbbVie and may hold AbbVie stock; **CL** has received honoraria for being on the speaker's bureau and advisory board for AbbVie.

These data were previously presented at the 18th World Congress of the International Society of Physical and Rehabilitation Medicine, Sydney, Australia, June 1–6, 2024. (Ref. DV-011154)

References
1. Wissel J et al. *J Pain Symptom Manage.* 2000;20(1):44-9.
2. Francisco GE et al. *J Rehabil Med.* 2017;49(8):659-66.
3. Botox Label [package insert]. Allergan USA; 2021.
4. Wissel J et al. *J Pain and Symptom Manage.* 2016;52(1):17-26.
5. Francisco GE et al. *PM&R.* 2020;12(11):1120-33.
6. Esquenazi A et al. *PM&R.* 2021;13(10):1079-1093.

INTRODUCTION

- Spasticity is a motor disorder characterized by muscle stiffness, tightness, and involuntary muscle contractions, which can lead to difficulties in movement and coordination and patients suffering from spasticity may also suffer from pain[1-2]
- OnabotulinumtoxinA (onabotA) is indicated for the treatment of upper limb (UL) and lower limb (LL) spasticity[3]
- Limited data exists to support the reduction of pain after onabotA treatment in patients with spasticity[4-6]; thus, a better understanding is needed on the effect of onabotA on pain in these patients
- OnabotA may be an effective treatment for spasticity patients with pain across age groups with any etiology

METHODS

Study Design and Patients

- The Adult Spasticity International Registry (ASPIRE) is an international, multicenter, prospective, observational study (NCT01930786), that examined onabotA utilization and effectiveness in treating adult patients with spasticity regardless of prior treatment with onabotA treatment
- Patients with spasticity across multiple etiologies were treated with onabotA at the clinician's discretion over 2 years
- Patients stratified based on statistical criteria as "< 55 years" and "≥ 55 years" of age and who received ≥ 1 onabotA treatment for spasticity and had pain (NPRS >0) at baseline are described in this subanalysis

Assessments

- OnabotA treatment utilization was evaluated over 8 treatment sessions
- Pain was assessed with the Numeric Pain Rating Scale (NPRS), changes in functional impairment was assessed by the Disability Assessment Scale (DAS) pain subscale for UL and LL, and patient and physician satisfaction was measured after each treatment session
- Safety was assessed throughout the 2-year study
- A mixed effects regression model was used to assess changes over time; p values and confidence intervals were adjusted for multiple comparisons using a multivariate t distribution

ASPIRE Data Collection

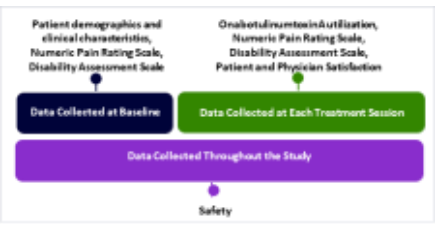
Patient demographics and clinical characteristics, Numeric Pain Rating Scale, Disability Assessment Scale
OnabotulinumtoxinA utilization, Numeric Pain Rating Scale, Disability Assessment Scale, Patient and Physician Satisfaction
Data Collected at Baseline
Data Collected at Each Treatment Session
Data Collected Throughout the Study
Safety

RESULTS

Baseline Characteristics

- Out of the 730 patients in ASPIRE who received ≥1 onabotulinumtoxinA (onabotA) treatment for spasticity, a total of 494 patients had pain (defined as NPRS > 0) at baseline
- The 494 spasticity patients with baseline NPRS >0 included in this subanalysis:
 - Were evenly distributed by age (< 55 years [n = 244, 49.3%] and ≥ 55 years [n = 250, 50.6%])
 - Were predominantly female (59% and 54%, respectively), largely White (71% and 81%, respectively), and 67.8% of patients from each age group received prior onabotA treatment for spasticity
 - Had stroke as the most frequently reported etiology (41% vs 70%, respectively)

Total OnabotulinumtoxinA Dose by Treatment Session

	Tx 1	Tx 2	Tx 3	Tx 4	Tx 5	Tx 6	Tx 7	Tx 8
Total Dose (U) in < 55 years	n = 244	n = 209	n = 173	n = 148	n = 112	n = 89	n = 57	n = 16
Mean (SD)	351.8 (200.8)	352.8 (198.6)	355.3 (197.3)	372.3 (209.0)	395.7 (209.8)	371.1 (194.4)	381.0 (187.8)	447.2 (243.3)
Min, Max	40.0, 1000.0	50.0, 1125.0	60.0, 1100.0	50.0, 1200.0	100.0, 1200.0	62.0, 1225.0	100.0, 1200.0	160.0, 1200.0
Total Dose (U) in ≥ 55 years	n = 250	n = 209	n = 176	n = 146	n = 111	n = 72	n = 47	n = 12
Mean (SD)	337.9 (191.9)	347.9 (173.1)	365.3 (165.7)	363.6 (169.4)	384.5 (154.5)	375.1 (149.9)	387.0 (138.3)	485.4 (131.0)
Min, Max	30.0, 1100.0	45.0, 930.0	50.0, 830.0	50.0, 800.0	50.0, 800.0	50.0, 650.0	50.0, 600.0	300.0, 710.0

n, number of patients in each treatment session by age group category; SD, standard deviation; Tx, treatment session; U, units

- The mean onabotA dose over the 8 treatment sessions ranged from 351.8 U to 447.2 U in patients < 55 years compared to 337.9 U to 485.4 U in patients ≥ 55 years

Patients in Both Age Groups Had Improvements in Pain From Baseline After OnabotulinumtoxinA Treatment as Assessed by the Numeric Pain Rating Scale (NPRS)

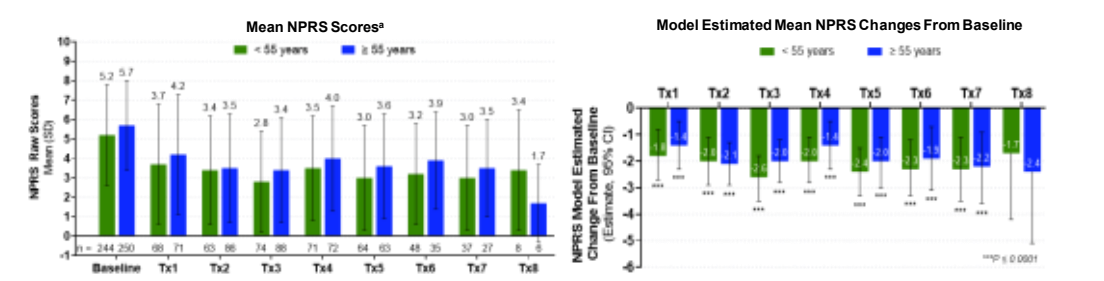
Mean NPRS Scores[a]
< 55 years
≥ 55 years
NPRS Raw Scores Mean (SD)
Baseline
Tx1
Tx2
Tx3
Tx4
Tx5
Tx6
Tx7
Tx8
Model Estimated Mean NPRS Changes From Baseline
NPRS Model Estimated Change From Baseline (Estimate, 95% CI)
***P ≤ 0.0001

NPRS, numeric pain rating scale; n, number of patients; Tx, treatment session. [a]NPRS rates the level of pain experienced in the last 24 hours from 0 (no pain) to 10 (highest pain imaginable). The baseline NPRS was evaluated at office visit 1; response to treatments (Tx) 2 - 8 were collected by phone or web after each treatment. The baseline NPRS reflects the initial measurements available for the cohort patients treated at any subsequent time point. Missing or ambiguous responses resulted in variation in the number of n's for each graph. A multivariate t distribution method is used to adjust confidence intervals and p values for multiple comparisons. *Indicates statistically significant differences between txs.

- Patients with spasticity in both age groups had improvements in pain from baseline
 - Mean NPRS scores for pain decreased from 5.2 at baseline to 3.4 at Tx 8 in patients < 55 years
 - Mean NPRS scores for pain decreased from 5.7 at baseline to 1.7 at Tx 8 in patients ≥ 55 years
- Model estimated NPRS was significantly reduced from baseline after Tx 1 - 7 for both age groups

Disability Assessment Scale (DAS)a Pain Subscale for UL and LL Spasticity Improved Relative to Baseline Across all Treatment Sessions

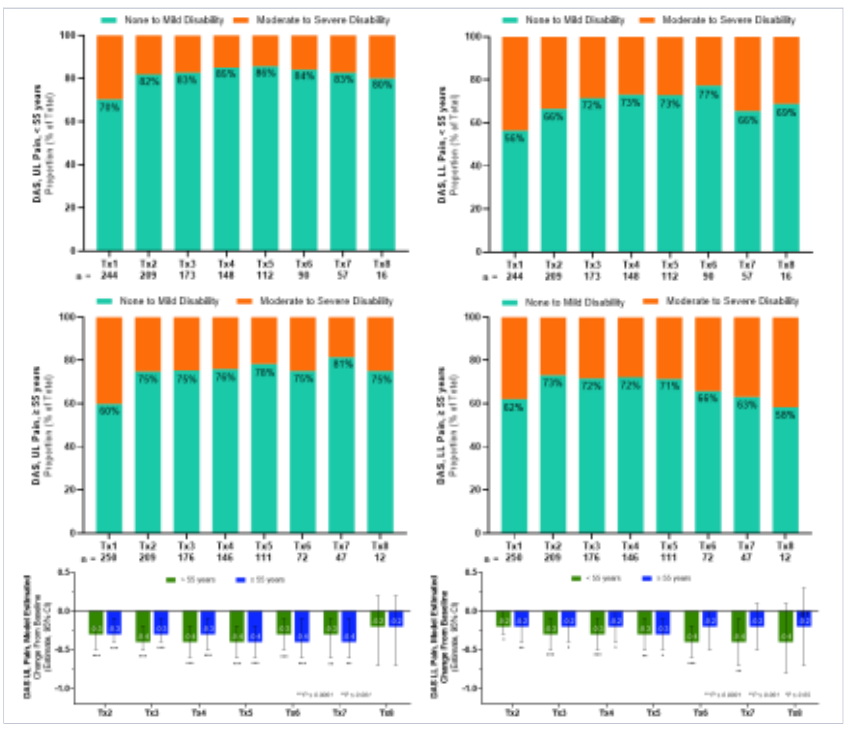
None to Mild Disability
Moderate to Severe Disability
DAS, UL Pain, < 55 years Proportion (% of Total)
DAS, LL Pain, < 55 years Proportion (% of Total)
DAS, UL Pain, ≥ 55 years Proportion (% of Total)
DAS, LL Pain, ≥ 55 years Proportion (% of Total)
< 55 years
≥ 55 years

DAS, disability assessment scale; n, number of patients; onabotA, onabotulinumtoxinA, UL, upper limb; LL, lower limb; Tx, treatment sessions. [a]DAS objectively evaluates functional impairment, such as pain, resulting from spasticity. DAS uses a 4-point rating scale, with 0 as no disability; 1, mild disability; 2, moderate disability; 3, severe disability; baseline DAS measurement was taken prior to injection with onabotulinumtoxinA at the first treatment visit (Tx 1), thus, Tx 1 represents the baseline level of disability. DAS for treatments (Tx) 2 – 8 reflects response to the prior treatment (ie, Tx 2 reflects the response to Tx 1). Missing or ambiguous responses resulted in variation in the number of n's for each panel. A multivariate t distribution method is used to adjust confidence intervals and p values for multiple comparisons. *Indicates statistically significant differences between treatments.

- The majority of spasticity patients in both groups (≥ 70% patients in the < 55 years group and ≥ 60% patients in the ≥ 55 years group) reported none to mild disability for UL
- A lower proportion of patients reported none to mild disability for LL compared to UL (≥ 56% patients in the <55 years and ≥ 58% patients in the ≥ 55 years groups)
- Model estimated DAS for UL spasticity on pain subscale was significantly reduced after onabotA treatment compared to baseline across most treatment sessions (Tx 2 – 7) regardless of age group
- Model estimated DAS for LL spasticity on pain subscale showed significant reduction in pain compared to baseline across most treatment sessions regardless of age group

Increased Patient Satisfaction After OnabotulinumtoxinA Treatments Across Age Groups

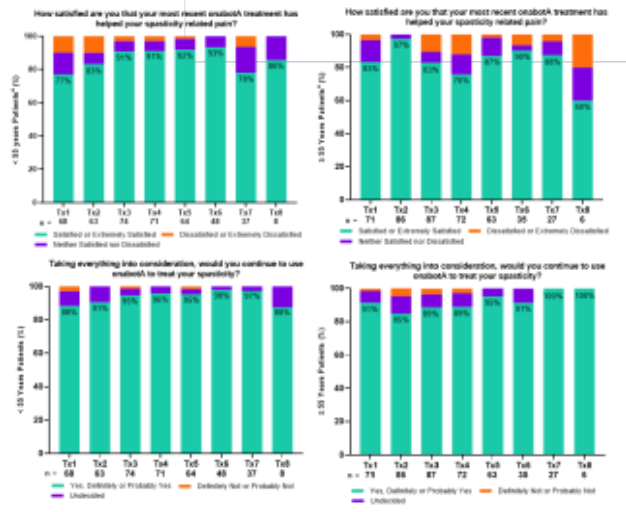
How satisfied are you that your most recent onabotA treatment has helped your spasticity related pain?
Satisfied or Extremely Satisfied
Dissatisfied or Extremely Dissatisfied
Neither Satisfied nor Dissatisfied
Taking everything into consideration, would you continue to use onabotA to treat your spasticity?
Yes, Definitely or Probably Yes
Definitely Not or Probably Not
Undecided

n, number of respondents; N/A, not applicable; onabotA, onabotulinumtoxinA; Tx, treatment session. [a]Not applicable (N/A) responses to this question (the only question for which N/A could have been selected) are not included in bars but are included in the total number of respondents below each treatment. Patient-reported outcomes were collected at baseline and approximately five weeks after each treatment; a follow-up interview was conducted 12 weeks after the final treatment. Missing or ambiguous responses resulted in variation in the number of n's for each panel.

- OnabotA treatment resulted in overall high patient satisfaction across age groups
 - After each treatment session, ≥ 77% of spasticity patients in the < 55 years group and ≥ 60% in the ≥ 55 years group were satisfied that onabotA treatment helped their pain
 - After each treatment session, comparable proportion of patients in both age groups (≥ 88% of patients in the < 55 years and ≥ 85% in the ≥ 55 years) considered continuing using onabotA to treat their spasticity

Increased Physician Satisfaction After OnabotulinumtoxinA Treatments Across Age Groups

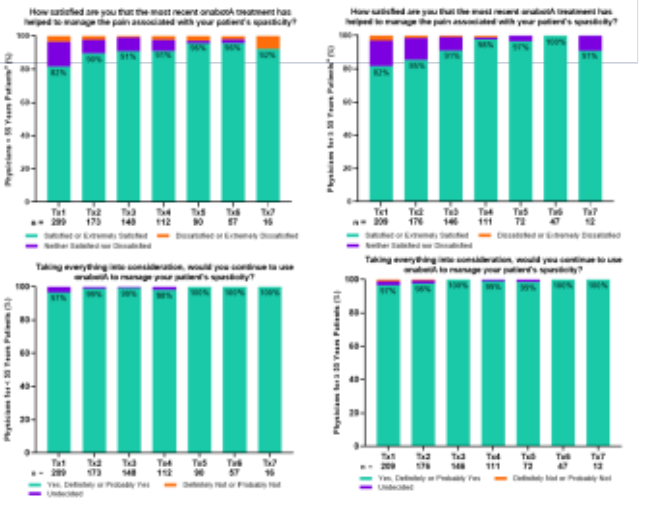
How satisfied are you that the most recent onabotA treatment has helped to manage the pain associated with your patient's spasticity?
Satisfied or Extremely Satisfied
Dissatisfied or Extremely Dissatisfied
Neither Satisfied nor Dissatisfied
Taking everything into consideration, would you continue to use onabotA to manage your patient's spasticity?
Yes, Definitely or Probably Yes
Definitely Not or Probably Not
Undecided

n, number of respondents; N/A, not applicable; Tx, treatment session. [a]Not applicable (N/A) responses to this question (the only question for which N/A could have been selected) are not included in bars but are included in the total number of respondents below each treatment. Physician-reported outcomes for treatments (Tx) 1 - 7 were collected at each office visit subsequent to treatment. Study requirements did not stipulate a final follow-up visit; thus, clinician-reported outcomes for Tx 8 were not evaluated. Missing or ambiguous responses resulted in variation in the number of n's for each panel.

- OnabotA treatment resulted in overall high physician satisfaction to treat their patient's spasticity across age groups
 - After each treatment session, ≥ 82% of physicians of spasticity patients in in both age groups were satisfied that the onabotA treatment helped their patient's pain
 - After each treatment session, ≥ 97% of physicians of spasticity patients in both age groups indicated they would continue to use onabotA to treat their patient's spasticity

Safety

- Overall, 230 nonserious adverse events (AEs) were reported in 86/244 patients (35.2%) in the < 55 years age group; 234 nonserious AEs were reported in 86/250 patients (34.4%) in the ≥ 55 years age group
 - 10 nonserious AEs in 10/244 patients (4.1%) in the < 55 years age group were considered treatment-related; the most common treatment-related AE was muscular weakness (3 events in 3/244 patients, 1.2%)
 - 7 nonserious AEs in 5/250 (2.0%) in the ≥ 55 years age group were considered treatment-related; the most common treatment-related AE was muscular weakness (3 events in 3/250 patients,1.2%)
- Overall, no serious AEs were reported in patients in the < 55 years age group; 3 serious AEs were reported in 2/250 (0.8%) patients in the ≥ 55 years age group and were considered treatment related: muscular weakness, dysphagia, and slow speech

Study Limitations

- Given the observational nature of the study, the generality of this analysis is limited
- Low sample sizes for some outcomes an treatment sessions (due to missing or ambiguous responses resulting in variation in the number of n's for each treatment session)
- DAS has been validated for UL spasticity only; however, it has also been used to evaluate LL spasticity

Presented at the Italian Society of Neurology (SIN) – 55° National Congress, October, 24-25, 2025- Padova, Italy. (CLR-004511)

24-28 Ottobre 2025
Padova Congress

55° CONGRESSO SOCIETÀ ITALIANA DI NEUROLOGIA