754

Pain in Patients With Spasticity: Patient Characteristics From the Adult Spasticity International Registry (ASPIRE) Study

Manuela Gorini,* Esther Duarte,[1] Jörg Wissel,[2] Cassandra List,[3] Marc Schwartz,[4] Mariana Nelson,[5] Tiziana Musacchio,[6] Wuwei Feng[7]

[1]Department of Physical Medicine and Rehabilitation, Hospitals del Mar i l'Esperança, Parc de Salut Mar, Barcelona, Spain; [2]Neurology and Psychosomatic at Wittenbergplatz, Berlin and University of Potsdam, Germany; [3]Brooks Rehabilitation, Jacksonville, FL, USA; [4]MS Biostatistics, LLC, Clermont, FL, USA; [5]AbbVie, Irvine, CA, USA; [6]AbbVie, Rome, Italy; *AbbVie srl Rome, Italy. This individual has been added to this publication author by-line, with the permission of the original authors, for the express purpose of conducting the presentation at a local congress or in a local language. She did not contribute to the content of the publication. [7]Department of Neurology, Duke University School of Medicine, Durham, NC, USA

OBJECTIVE

To describe the baseline demographics and clinical characteristics in spasticity patients from any etiology and pain (NPRS > 0) at baseline using data from the ASPIRE study

CONCLUSIONS

Real-world baseline characteristics of spasticity patients of any etiology and pain at baseline (NPRS > 0) provide important information about pain in patients seeking onabotulinumtoxinA treatment for spasticity

The effectiveness and safety of onabotulinumtoxinA in this patient population continues to be evaluated

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial. AbbVie funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Gina E. Elsen, PhD, of AbbVie. Editorial assistance was provided by Angela T. Hadsell, of AbbVie. Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: Esther Duarte has received fees for study participation from Allergan, Dysport, and Merz; Jörg Wissel has received honoraria for being on the speaker's bureaus and advisory boards for AbbVie, Ipsen, Medtronic, and Merz; Cassandra List has received honoraria for being on the speaker's bureau and advisory board for AbbVie; Marc Schwartz is a biostatistical consultant for AbbVie; Mariana Nelson, Tiziana Musacchio and Manuela Gorini are full-time employees of AbbVie and may hold AbbVie stock; Wuwei Feng has received research grants from the National Institutes of Health (NIH) and AHA.

These data were previously presented at World Federation for Neurorehabilitation (WFNR) 2024, Vancouver, Canada (Ref. DV-011147)

INTRODUCTION

- Spasticity is a disorder characterized by muscle stiffness, tightness, and involuntary muscle contractions, which can lead to difficulties in movement and coordination, and patients suffering from spasticity may also suffer from pain[1-2]
- OnabotulinumtoxinA (onabotA) is indicated for treatment of upper limb (UL) and lower limb (LL) spasticity[3]
- Limited data is available assessing pain management in adults with spasticity[4-6]
- A better understanding of the effectiveness of onabotA on pain in patients with spasticity is needed to inform treatment decisions in clinical practice

ASPIRE, Adult Spasticity international Registry; NPRS, numeric pain rating scale.[1]Wissel J et al. J Pain Symptom Manage. 2000;20(1):44-9; [2]Francisco GE et al. J Rehabil Med. 2017;49(8):659-66; [3]Botox Label [package insert]. Allergan USA; 2021; [4]Wissel J et al. J Pain and Symptom Manage. 2016;52(1):17-26; [5]Francisco GE et al. PM&R. 2020;12(11):1120-33; [6]Esquenazi A et al. PM&R. 2021;13(10):1079-1093.

METHODS

Study Design

- The Adult Spasticity International Registry (ASPIRE) is an international, multicenter, prospective, observational study (NCT01930786) that examined onabotulinumtoxinA (onabotA) utilization and effectiveness in spasticity patients over a 2-year period
- Data from spasticity patients across etiologies treated with onabotA were analyzed
- This analysis includes spasticity patients who received with ≥ 1 onabotA treatment and who had pain (NPRS > 0) at baseline

Assessments at Baseline

- Patient demographics and clinical characteristics
- Pain was assessed with:

-Numeric Pain Rating Scale (NPRS)
-Disability Assessment Scale (DAS)

ASPIRE, Adult SPasticity International REgistry; DAS, disability assessment scale; NPRS, numeric pain rating scale; onabotA, onabotulinumtoxinA.

ASPIRE Data Collection During the Study Period

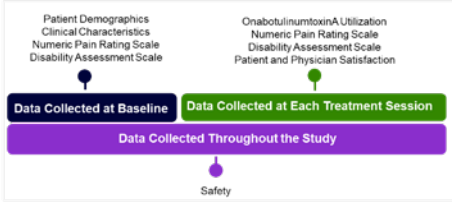
Patient Demographics
Clinical Characteristics
Numeric Pain Rating Scale
Disability Assessment Scale
OnabotulinumtoxinA Utilization
Numeric Pain Rating Scale
Disability Assessment Scale
Patient and Physician Satisfaction
Data Collected at Baseline
Data Collected at Each Treatment Session
Data Collected Throughout the Study
Safety

RESULTS

Out of 730 Patients in ASPIRE Who Received ≥ 1 OnabotA for Spasticity, 68% Reported Pain at Baseline (NPRS > 0) and Were Included in This Analysis - Patient Demographics

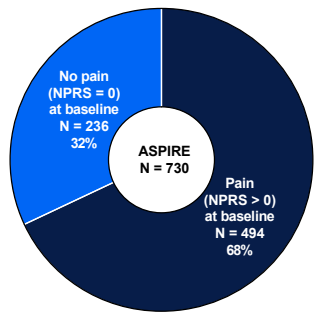
No pain
(NPRS = 0)
at baseline
N = 236
32%
ASPIRE
N = 730
Pain
(NPRS > 0)
at baseline
N = 494
68%

Parameter	Patients with NPRS > 0 N = 494
Age, years, mean (SD)	54.0 (15.4)
Sex, n (%)	
Female	281 (56.9%)
Male	213 (43.1%)
Race, n (%)	
American Indian or Alaska Native	1 (0.2%)
Asian	34 (6.9%)
Black or African American	59 (11.9%)
Latino/Hispanic	13 (2.6%)
Middle Eastern or Arabic	3 (0.6%)
White	375 (75.9%)
Data not available	8 (1.6%)
Other	1 (0.2%)
BMI, m², mean (SD)[a]	26.7 (5.8)

ASPIRE, Adult SPasticity International REgistry; BMI, body mass index; n, number of patients; NPRS, numeric pain rating scale; onabotA, onabotulinumtoxinA; SD, standard deviation. [a]BMI data for 401 patients were included in this analysis; percentage was calculated using this number of patients.

Most Patients (62%) Received Prior OnabotA Treatment for Spasticity

- Mean time from prior onabotA treatment for spasticity was 7.7 months

Clinical Characteristics

Parameter	Patients with NPRS > 0 N = 494
Prior onabotA treatment for spasticity, (%)	
Yes	305 (61.7%)
No	189 (38.3%)
Time from prior onabotA treatment for spasticity, months[a]	
Mean (SD)	7.7 (14.9)
Min, Max	0.1, 186.4
OnabotA treatment within the past 12 weeks, n (%)	
No	488 (98.8%)
Yes	6 (1.2%)

NPRS, numeric pain rating scale; OnabotA, onabotulinumtoxinA; SD, standard deviation. [a] Patients with NPRS > 0 at baseline; [b] Data from 294 patients (with NPRS > 0 at baseline) were included in this analysis; percentage was calculated using this number of patients

Stroke Was the Most Frequently Reported Etiology[a] at Baseline in 56% of Spasticity Patients With Pain (NPRS[b] > 0) at Baseline

Etiology in Patients With Spasticity and Pain at Baseline (N = 494)

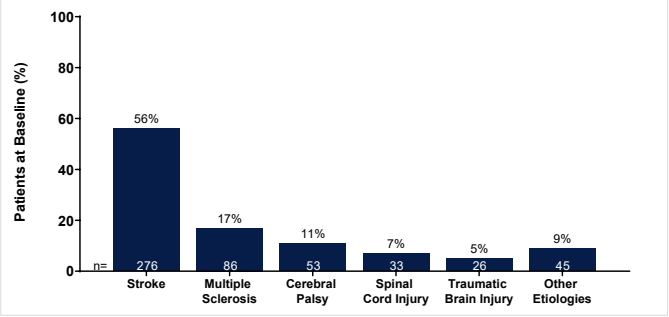
Patients at Baseline (%)
100
80
60
40
20
0
n=
56%
276
Stroke
17%
86
Multiple Sclerosis
11%
53
Cerebral Palsy
7%
33
Spinal Cord Injury
5%
26
Traumatic Brain Injury
9%
45
Other Etiologies

NPRS, numeric pain rating scale; n, number of patients. [a] Patients may have more than one etiology; [b] NPRS scale: 0 (no pain) to 10 (worst pain imaginable).

Baseline Clinical Presentation in Spasticity Patients With Pain at Baseline - Upper and Lower Limb Spasticity[a,b]

Most Common Upper Limb Spasticity Presentations at Baseline Were Clenched Fist (63%) and Flexed Elbow (62%)

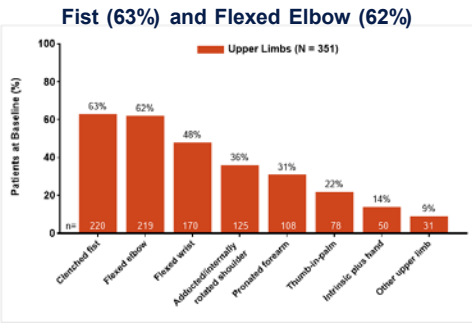
Upper Limbs (N = 351)
Patients at Baseline (%)
63%
220
Clenched fist
62%
219
Flexed elbow
48%
170
Flexed wrist
36%
125
Adducted/internally rotated shoulder
31%
108
Pronated forearm
22%
78
Thumb-in-palm
14%
50
Intrinsic plus hand
9%
31
Other upper limb

Most Common Lower Limb Spasticity Presentation at Baseline Was Equinovarus Foot (72%)

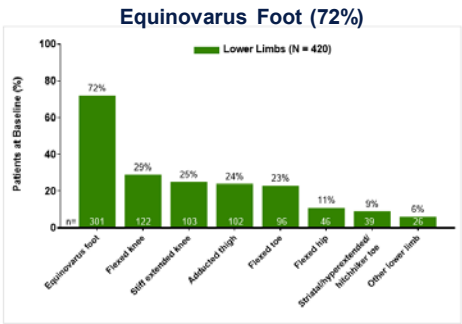
Lower Limbs (N = 420)
Patients at Baseline (%)
72%
301
Equinovarus foot
29%
122
Flexed knee
25%
103
Stiff extended knee
24%
102
Adducted thigh
23%
96
Flexed toe
11%
46
Flexed hip
9%
39
Striatal/hyperextended/ hitchhiker toe
6%
26
Other lower limb

ASPIRE, Adult SPasticity International Registry; n, number of patients.[a] Patients may not have been treated during ASPIRE; [b] Percentages may add to >100% as patients may have more than one presentation.

Most Patients[a] (40%) Were Treated for Both Upper and Lower Limb Spasticity

- Among the 494 spasticity patients[a] with pain (NPRS > 0) at baseline, 122 (25%) were treated for upper limb, 172 (35%) were treated for lower limb, and 200 (40%) were treated for upper and lower limb spasticity

Limbs Treated at Baseline

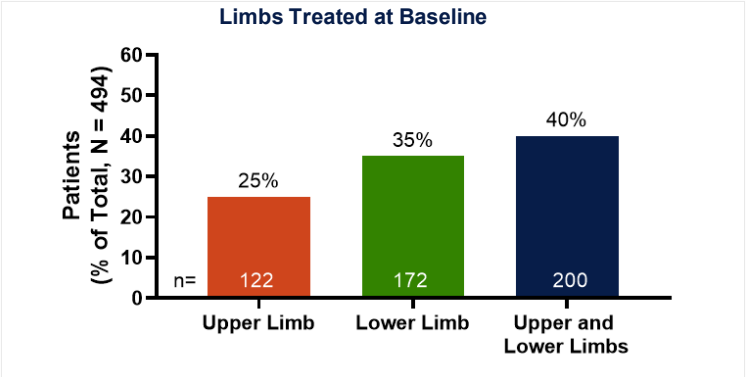
Patients
(% of Total, N = 494)
60
50
40
30
20
10
0
n=
25%
122
Upper Limb
35%
172
Lower Limb
40%
200
Upper and Lower Limbs

ASPIRE, Adult SPasticity International Registry; NPRS, numerical pain rating scale; n, number of patients. [a] Patients may not have been treated during ASPIRE.

Mean Baseline Pain Score Assessed With NPRS Was 5.4

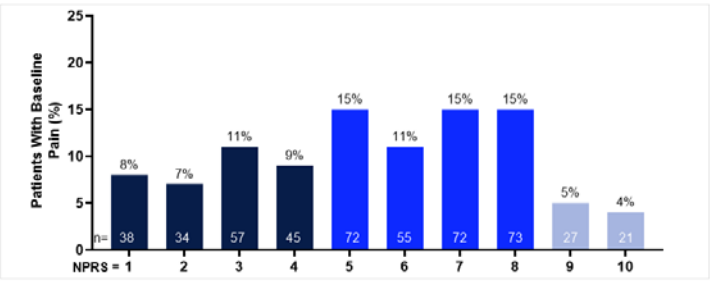
Patients With Baseline Pain (%)
25
20
15
10
5
0
n=
NPRS = 1
8%
38
2
7%
34
3
11%
57
4
9%
45
5
15%
72
6
11%
55
7
15%
72
8
15%
73
9
5%
27
10
4%
21

	Upper Limb (n = 122)	Lower Limbs (n = 172)	Upper and Lower Limbs (n = 200)	Total (N = 494)
NPRS				
Mean (SD)	**5.3 (2.5)**	**5.4 (2.4)**	**5.6 (2.5)**	**5.4 (2.5)**
Min, Max	1.0, 10.0	1.0, 10.0	1.0, 10.0	1.0, 10.0
Median [IQR]	5.0 [3.0, 7.0]	6.0 [3.0, 7.0]	6. 0 [4.0, 8.0]	6.0 [3.0, 7.0]

IQR, inter-quartile range; n, number of patients; NPRS, numeric pain rating scale; SD, standard deviation.[a] Patients may not have been treated during ASPIRE; [b] NPRS is a numeric scale in which a respondent selects a whole number from 0 (no pain) to 10 (worst pain imaginable) that best reflects the intensity of their pain; spasticity patients included in this subanalysis had NPRS > 0.

DAS[a] Pain Subscale Levels at Baseline Were Reported as None or Mild by 65% (UL, n = 319) and 59% (LL, n = 292) of Patients

DAS Upper and Lower Limb Pain Subscale Levels at Baseline

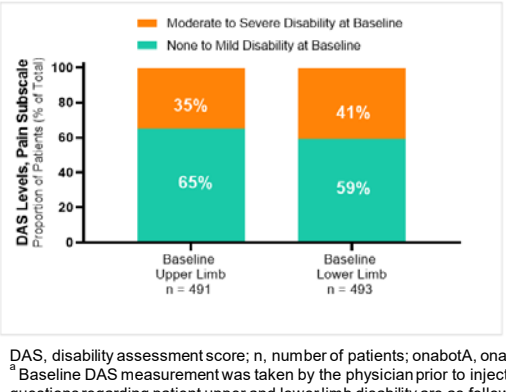
Moderate to Severe Disability at Baseline
None to Mild Disability at Baseline
DAS Levels, Pain Subscale
Proportion of Patients (% of Total)
100
80
60
40
20
0
35%
65%
Baseline
Upper Limb
n = 491
41%
59%
Baseline
Lower Limb
n = 493

DAS Upper and Lower Limb Pain Levels and Scores[b] at Baseline

	Upper Limb Patients	Lower Limb Patients
DAS baseline pain scores		
Mean (SD)	**1.1** (1.1)	**1.2** (1.1)
Min, Max	0.0, 3.0	0.0, 3.0
Median [IQR]	1.0 [0.0, 2.0]	1.0 [0.0, 2.0]
DAS baseline pain subscale levels, n (%)	**n = 491[c]**	**n = 493[c]**
None disability	201 (40.9)	160 (32.5)
Mild disability	118 (24.0)	132 (26.8)
Moderate disability	108 (22.0)	128 (6.0)
Severe disability	64 (13.0)	73 (14.8)

DAS, disability assessment score; n, number of patients; onabotA, onabotulinumtoxinA; UL, upper limb; LL, lower limb; SD, standard deviation. [a] Baseline DAS measurement was taken by the physician prior to injection with onabotA at the first treatment visit (Tx1); [b] Scoring for the questions regarding patient upper and lower limb disability are as follows: 0 = none disability, 1 = mild disability, 2 = moderate disability, 3 = severe disability; [c] Number of patients assessed at baseline.

Limitations:

- Given the observational nature of the study, the generality of this analysis is limited
- Low sample sizes for some outcomes and treatment sessions (due to missing or ambiguous responses resulting in variation in the number of n's for each treatment session)
- DAS has been validated for UL spasticity only; however, it has also been used to evaluate LL spasticity

Presented at the Italian Society of Neurology (SIN) – 55° National Congress, October, 24-25, 2025- Padova, Italy. (CLR-004510)

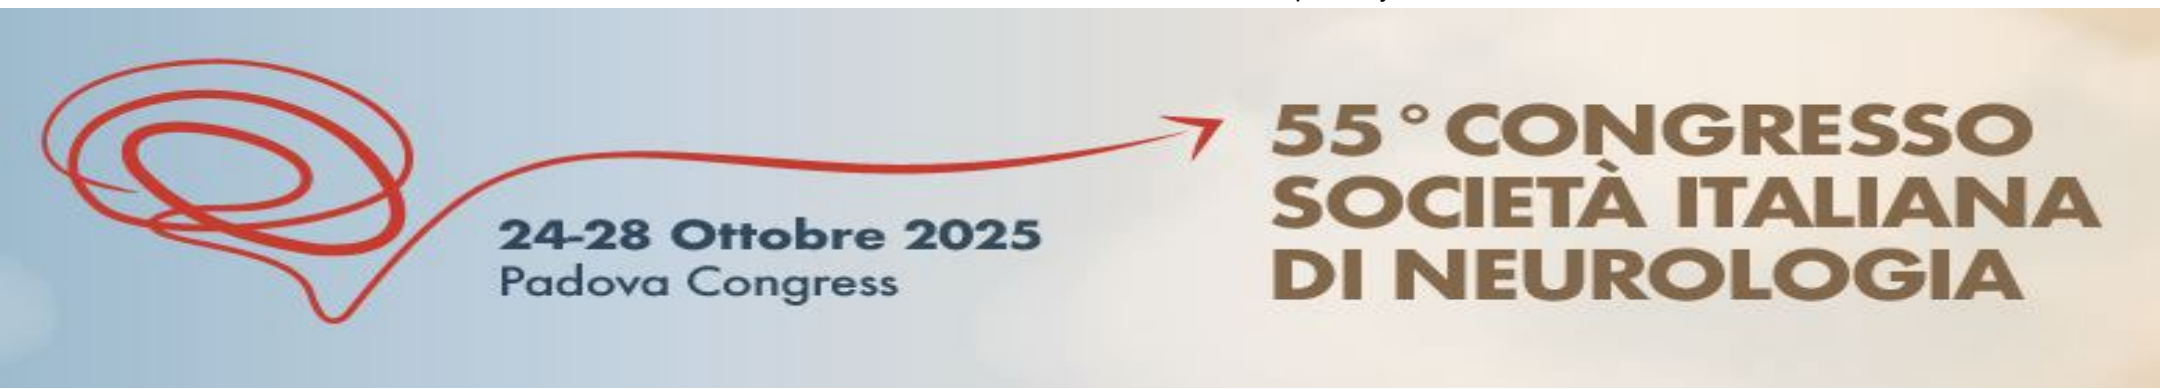
24-28 Ottobre 2025
Padova Congress
55° CONGRESSO
SOCIETÀ ITALIANA
DI NEUROLOGIA