833

Cluster Analysis of Patient Characteristics Associated with Up to 100% "Off" Time Improvement from Two Phase 3 Trials

Ilenia De Ionna,* Khashayar Dashtipour,[1] Rajesh Pahwa,[2] Bruno Bergmans,[3] Per Odin,[4] Megha Shah,[5] Lars Bergmann,[5] Jaclyn Homola,[5] Connie H. Yan,[5] Resmi Gupta,[5] Michael J. Soileau[6]

*[1]Loma Linda University Health System, Loma Linda, CA, USA; [2]University of Kansas Medical Center, Kansas City, KS, USA; [3]AZ St-Jan Brugge, Brugge, Belgium and Ghent University Hospital, Ghent, Belgium; [4]Skåne University Hospital, Lund University, Lund, Sweden; [5]AbbVie Inc., North Chicago, IL, USA; *AbbVie srl Rome, Italy. This individual has been added to this publication author by-line, with the permission of the original authors, for the express purpose of conducting the presentation at a local congress or in a local language. She did not contribute to the content of the publication. [6]Texas Movement Disorder Specialists, Georgetown, TX, USA*

OBJECTIVE

To evaluate post hoc the baseline characteristics of patients with advanced Parkinson's disease (PD) from two phase 3 clinical trials who received 24-hour/day continuous subcutaneous infusion (CSCI) therapy of foslevodopa/foscarbidopa (LDp/CDp) and achieved up to 100% improvement in "Off" time responses

CONCLUSIONS

This cluster analysis identified patient profiles with "Off" time improvement up to 100%, and those with the greatest improvement were characterised by less PD duration (since onset), less levodopa equivalent daily dose (LEDD), and less "Off" time hours at baseline

The patients with 100% "Off" time improvement were the largest cluster and approximately 50% of which had a baseline PD duration of < 10 years as well as a baseline "Off" time of < 5 hours/day

These post hoc data highlight the potential for personalised approaches to optimise outcomes with LDp/CDp CSCI therapy, and support the consideration of using LDp/CDp earlier in the advanced PD treatment algorithm; even as a viable first option for patients when symptoms are uncontrolled on oral therapy

AbbVie and the authors thank the patients, caregivers, study site personnel, and trial investigators who participated in these clinical trials. AbbVie Inc funded this study and participated in the study design; study research; collection, analysis, and interpretation of data; and writing, reviewing, and approving this publication. All authors had access to relevant data; participated in the drafting, review, and approval of this publication; and agreed to the submission of this abstract to the 11th Congress of the European Academy of Neurology (EAN-2025). No honoraria or payments were made for authorship. Matthew R. Distasi, of AbbVie Inc, provided medical writing support and Catherine Barone, of AbbVie Inc, provided editorial support during the development of this publication. Conflicts of Interest (Disclosures): AbbVie Inc supported this analysis. Financial relationships, activities, and/or interests (currently or within the past 2 years) of the authors with companies whose products may be related to the present report are listed as declared by the authors: KD received advisor/consulting &/or speaker fees from AbbVie Inc, Acadia, Allergan, Amneal, Ipsen, Merz, Neurocrine, Sunovian, Supernus, & Teva. RP is an AbbVie Inc investigator & has received consulting &/or research support from Abbott, AbbVie Inc, ACADIA, Acorda, Adamas, Addex, Amneal, Biogen, Biohaven, Boston Scientific, CalaHealth, EIP, Global Kinetics, Impax, Impel, Intec, Kyowa, Lilly, Lundbeck, Mitsubishi, Neuraly, Neurocrine, Neuroderm, Neuropharma, Orbis,Parkinson's Foundation, Pharma 2B, PhotoPharmics, Prilenia, Roche, SIS, Sun Pharma, Sunovion, Teva, Theranexus, Theravance, US WorldMeds, & Voyager. BB has received advisor/speaker fees &/or grants from AbbVie Inc, EG, Ipsen, Merz, & Zambon. PO received: advisor/consultant &/or speaker fees from AbbVie Inc, Bial, Britannia, EVER Pharma, Nordic Infucare, STADA, & Zambon; and, PO's institution received research support from AbbVie Inc, Lund University Medical Faculty, Parkinsonfonden, SFO Multipark, Swedish Parkinson Academy, Swedish Research Council, & Health Care Region Skåne. MS, LB, JH, CHY, & RG are full-time employees of AbbVie Inc & may hold AbbVie Inc stock/stock options. MJS received advisor/consulting/speaker fees &/or research/ grant support from Abbott, AbbVie Inc, Amneal, Cerevel, CND Lifesciences, the HDSA, Jazz, Kyowa, Merz, Medtronic, Neurocrine, Praxis, Scion, Supernus, & Teva.
These data were previously presented , at the 11th Congress of the European Academy of Neurology (EAN-2025), June 21 – 24, 2025, Helsinki, Finland; (DV-016011)

INTRODUCTION

- As Parkinson's disease (PD) progresses, patients on oral levodopa (LD) medication experience motor fluctuations at varying rates due to reduced dopaminergic neuron buffering capacity as well as inconsistent oral LD absorption secondary to reduced gastric motility; resulting in fluctuations in plasma LD concentrations[1]
- Due to the heterogeneous nature and progression of PD, treatment responses also vary among patients[2]
- Foslevodopa/foscarbidopa (LDp/CDp) is a soluble formulation of levodopa/carbidopa (LD/CD) prodrugs, administered as a 24-hour/day continuous subcutaneous infusion (CSCI), which has demonstrated significant and sustained "Off" time reductions as well as a benefit-risk profile that was considered favourable in randomised controlled and open-label phase 3 trials[1,3]
- This analysis aimed to explore the baseline characteristics of patients from the active-controlled and open-label phase 3 LDp/CDp clinical trials who achieved "Off" time responses, including up to 100% improvement

METHODS

- This post hoc analysis examined the pooled baseline demographics and clinical characteristics from two phase 3 clinical trials of patients with advanced PD who received CSCI of LDp/CDp for 24 hours/day, including:
 - a 12-week, active-controlled, double-blind, randomised trial comparing the efficacy of LDp/CDp infusion vs oral LD/CD immediate-release (LD/CD-IR) therapies (NCT04380142)[1]
 - a 52-week, open-label trial evaluating the safety and efficacy of LDp/CDp infusion (NCT03781167)[3]
- The study designs, patient eligibility criteria, and primary results of both the 12-week, active-controlled as well as the 52-week, open-label clinical trials have been previously published[1,3]
- Patients with non-missing "Off" time data at baseline and at Week 12 or Week 13 in their Hauser diaries (based on the prespecified assessment time points in the active-controlled and open-label trials, respectively) were grouped into clusters based on their "Off" time improvement ranging from 0%–100%
- No statistical comparisons were performed in this exploratory analysis

RESULTS

Baseline Demographics and Clinical Characteristics in the Pooled Active-Controlled and Open-Label LDp/CDp-Treated Analysis Population Based on Patient Diary Reported "Off" Time Percent Improvement[a]

Parameters, N=158 (n [%])	0%[b] Improvement (30 [19.0])	1 – 25% Improvement (11 [7.0])	26 – 50% Improvement (22 [13.9])	51 – 75% Improvement (29 [18.4])	76 – 99% Improvement (24 [15.2])	100% Improvement (42 [26.6])
Sex, n (%)						
Female	11 (36.7)	7 (63.6)	5 (22.7)	14 (48.3)	12 (50.0)	10 (23.8)
Male	19 (63.3)	4 (36.4)	17 (77.3)	15 (51.7)	12 (50.0)	32 (76.2)
Age Category, n (%)						
< 50 years	4 (13.3)	0 (0)	0 (0)	2 (6.9)	1 (4.2)	4 (9.5)
50 – < 60 years	4 (13.3)	2 (18.2)	2 (9.1)	6 (20.7)	8 (33.3)	11 (26.2)
>= 60 – 80 years	21 (70.0)	8 (72.7)	20 (90.9)	19 (65.5)	15 (62.5)	27 (64.3)
> 80 years	1 (3.3)	1 (9.1)	0 (0)	2 (6.9)	0 (0)	0 (0)
PD Duration since Onset, n (%)						
< 10 years	12 (40.0)	6 (54.5)	7 (31.8)	7 (24.1)	12 (50.0)	22 (52.4)
10 – 14 years	11 (36.7)	4 (36.4)	7 (31.8)	13 (44.8)	7 (29.2)	11 (26.2)
> 14 years	7 (23.3)	1 (9.1)	8 (36.4)	9 (31.0)	5 (20.8)	9 (21.4)
LEDD, n (%)						
< 1800 mg	19 (63.3)	10 (90.9)	19 (86.4)	21 (72.4)	22 (91.7)	40 (95.2)
>= 1800 mg	11 (36.7)	1 (9.1)	3 (13.6)	8 (27.6)	2 (8.3)	2 (4.8)
Baseline "Off" Time, n (%)						
< 5 hours	12 (40.0)	2 (18.2)	4 (18.2)	9 (31.0)	4 (16.7)	20 (47.6)
5 – 7 hours	9 (30.0)	3 (27.3)	12 (54.5)	11 (37.9)	11 (45.8)	13 (31.0)
> 7 hours	9 (30.0)	6 (54.5)	6 (27.3)	9 (31.0)	9 (37.5)	9 (21.4)
Baseline "On" Time with nTSD, hours, mean (SD)[c]	2.80 (2.50)	2.67 (2.25)	3.13 (3.09)	2.78 (2.49)	1.88 (2.66)	2.56 (3.02)
Baseline "On" Time with TSD, hours, mean (SD)[c]	0.82 (1.41)	0.60 (0.85)	0.64 (1.00)	0.88 (1.55)	0.67 (1.31)	0.49 (0.94)

Abbreviations: %, percent; CDp, foscarbidopa; LDp, foslevodopa; LEDD, levodopa equivalent daily dose; N, total number of patients in the analysis population; n, number of patients included in the analysis subgroup or parameter; nTSD, non-Troublesome Dyskinesia; PD, Parkinson's disease; TSD, Troublesome Dyskinesia; SD, standard deviation

[a] Only patients with non-missing values for both the Baseline and Week 12 (in the Active-Controlled Trial) or Week 13 (in the Open-Label Trial) were included in the calculation of "Off" time percent improvement, based on the prespecified study visit and assessment schedule (ie, time points) in each trial:
[b] Patients in the "0% Improvement" subgroup demonstrated either no improvement or worsening:
[c] Average normalised values

The Groups/Clusters in the "Off" Time Improvement Patient Population

- Cluster analysis in the baseline population of N=158[a] patients showed the largest cluster corresponded to those with 100% improvement (27% of patients)

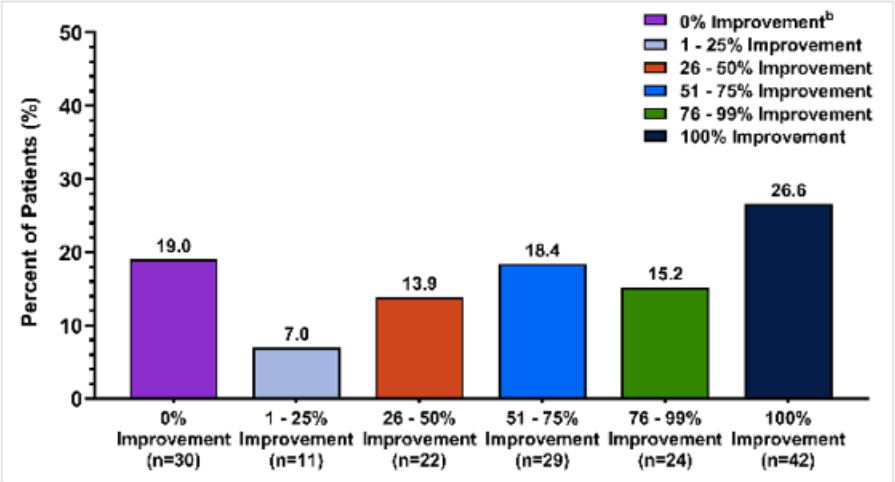
0% Improvement[b]
1 - 25% Improvement
26 - 50% Improvement
51 - 75% Improvement
76 - 99% Improvement
100% Improvement
Percent of Patients (%)
19.0
7.0
13.9
18.4
15.2
26.6
0% Improvement (n=30)
1 - 25% Improvement (n=11)
26 - 50% Improvement (n=22)
51 - 75% Improvement (n=29)
76 - 99% Improvement (n=24)
100% Improvement (n=42)

Abbreviations: %, percent; N, total number of patients in the analysis population; n, number of patients included in the analysis subgroup or cluster

[a] Only patients with non-missing values for both the Baseline and Week 12 (in the Active-Controlled Trial) or Week 13 (in the Open-Label Trial) were included in the calculation of "Off" Time percent improvement, based on the prespecified study visit and assessment schedule (ie, time points) in each trial
[b] Patients in the "0% Improvement" subgroup demonstrated either no improvement or worsening

Age Categories and LEDD in the "Off" Time Improvement Patient Population

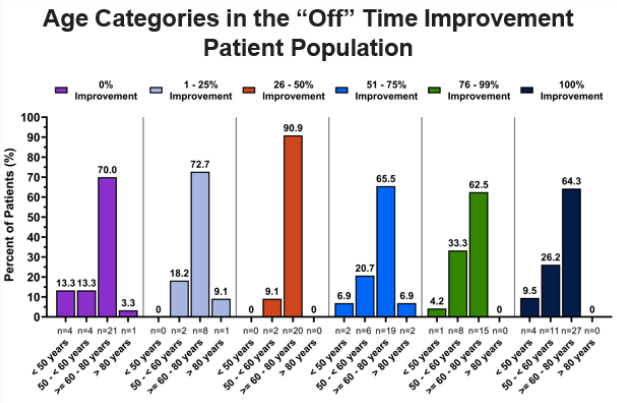
Age Categories in the "Off" Time Improvement Patient Population
0% Improvement
1 - 25% Improvement
26 - 50% Improvement
51 - 75% Improvement
76 - 99% Improvement
100% Improvement
Percent of Patients (%)
13.3 13.3 70.0 3.3
0 18.2 72.7 9.1
0 9.1 90.9 0
6.9 20.7 65.5 6.9
4.2 33.3 62.5 0
9.5 26.2 64.3 0
n=4 n=4 n=21 n=1
n=0 n=2 n=8 n=1
n=0 n=2 n=20 n=0
n=2 n=6 n=19 n=2
n=1 n=8 n=15 n=0
n=4 n=11 n=27 n=0
< 50 years
50 - < 60 years
>= 60 - 80 years
> 80 years

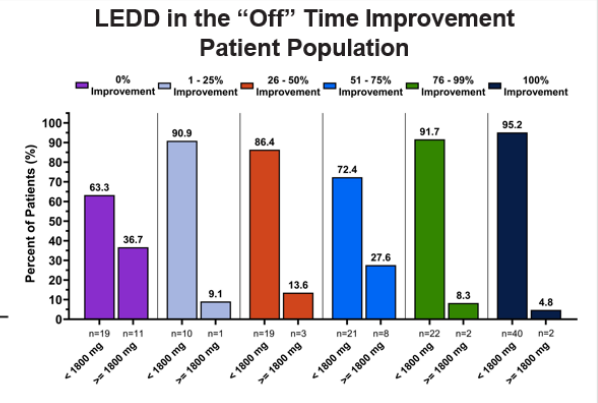
LEDD in the "Off" Time Improvement Patient Population
0% Improvement
1 - 25% Improvement
26 - 50% Improvement
51 - 75% Improvement
76 - 99% Improvement
100% Improvement
Percent of Patients (%)
63.3 36.7
90.9 9.1
86.4 13.6
72.4 27.6
91.7 8.3
95.2 4.8
n=19 n=11
n=10 n=1
n=19 n=3
n=21 n=8
n=22 n=2
n=40 n=2
< 1800 mg
>= 1800 mg

Abbreviations: %, percent; LEDD, levodopa equivalent daily dose; n, number of patients included in the analysis subgroup or cluster

- Across clusters, most patients were aged >= 60–80 years with < 1800 mg levodopa equivalent daily dose

PD Duration Since Onset and "Off" Time Hours per Day in the "Off" Time Improvement Patient Population

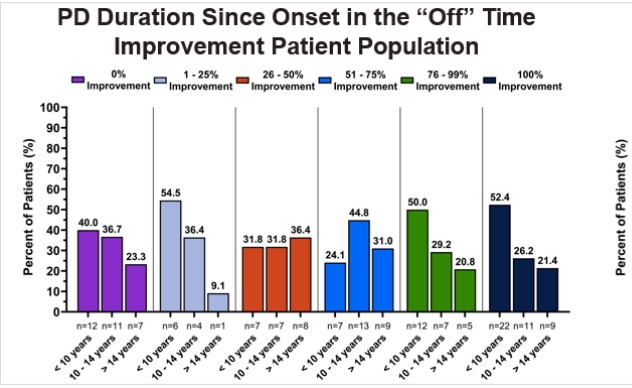
PD Duration Since Onset in the "Off" Time Improvement Patient Population
0% Improvement
1 - 25% Improvement
26 - 50% Improvement
51 - 75% Improvement
76 - 99% Improvement
100% Improvement
Percent of Patients (%)
40.0 36.7 23.3
54.5 36.4 9.1
31.8 31.8 36.4
24.1 44.8 31.0
50.0 29.2 20.8
52.4 26.2 21.4
n=12 n=11 n=7
n=6 n=4 n=1
n=7 n=7 n=8
n=7 n=13 n=9
n=12 n=7 n=5
n=22 n=11 n=9
< 10 years
10 - 14 years
> 14 years

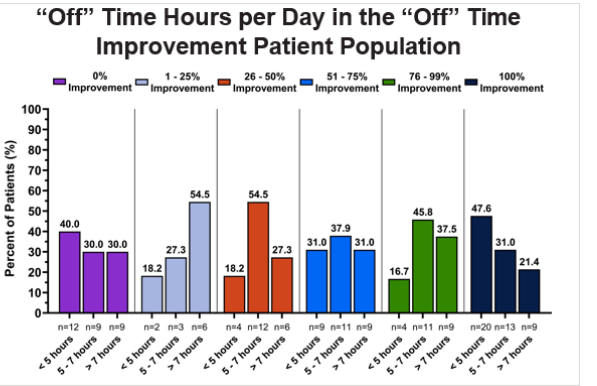
"Off" Time Hours per Day in the "Off" Time Improvement Patient Population
0% Improvement
1 - 25% Improvement
26 - 50% Improvement
51 - 75% Improvement
76 - 99% Improvement
100% Improvement
Percent of Patients (%)
40.0 30.0 30.0
18.2 27.3 54.5
18.2 54.5 27.3
31.0 37.9 31.0
16.7 45.8 37.5
47.6 31.0 21.4
n=12 n=9 n=9
n=2 n=3 n=6
n=4 n=12 n=6
n=9 n=11 n=9
n=4 n=11 n=9
n=20 n=13 n=9
< 5 hours
5 - 7 hours
> 7 hours

Abbreviations: %, percent; n, number of patients included in the analysis subgroup or cluster; PD, Parkinson's disease

- The 100% improvement cluster was characterised by patients with < 10 years since PD onset (ie, PD duration) and < 5 hours/day "Off" time at baseline

References

1. Aldred J, et al. *Neurol Ther.* 2023;12(6):1937-1958.
2. Hähnel T, et al. *npj Parkinsons Dis.* 2024;10:95.
3. Soileau MJ, et al. *Lancet Neurol.* 2022;21(12):1099-1109.

Presented at the Italian Society of Neurology (SIN) – 55° National Congress, October, 24-25, 2025- Padova, Italy. (CLR-004774)

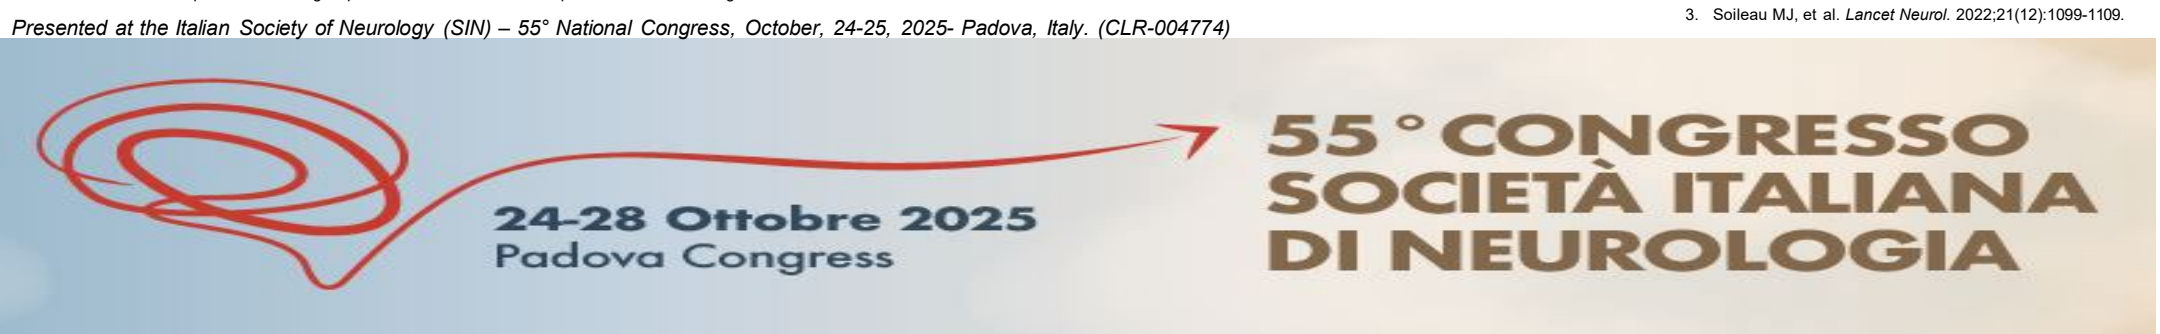
24-28 Ottobre 2025
Padova Congress
55° CONGRESSO
SOCIETÀ ITALIANA
DI NEUROLOGIA